A Phase Function to Find Periodicity in Spike Time Sequences - Application to Globus Pallidus neurons

UTSA Neurosciences Institute

Ramana Dodla and Charles J. Wilson ✶ Department of Biology, University of Texas at San Antonio, San Antonio, TX

Main points:

- ✶ Devise a better method to find correlations in short spike time data.
- ✶ Compare the method with correlation function.
- ✶ Apply the method to recordings of Globus Pallidus (GPe) neurons that are known to exhibit spontaneous but irregular oscillations.
- ✶ Examine whether periodicity is inherent in the random looking spike trains.
- ✶ Remove the periodic component, and characterize the underlying noise.

Phase function construction:

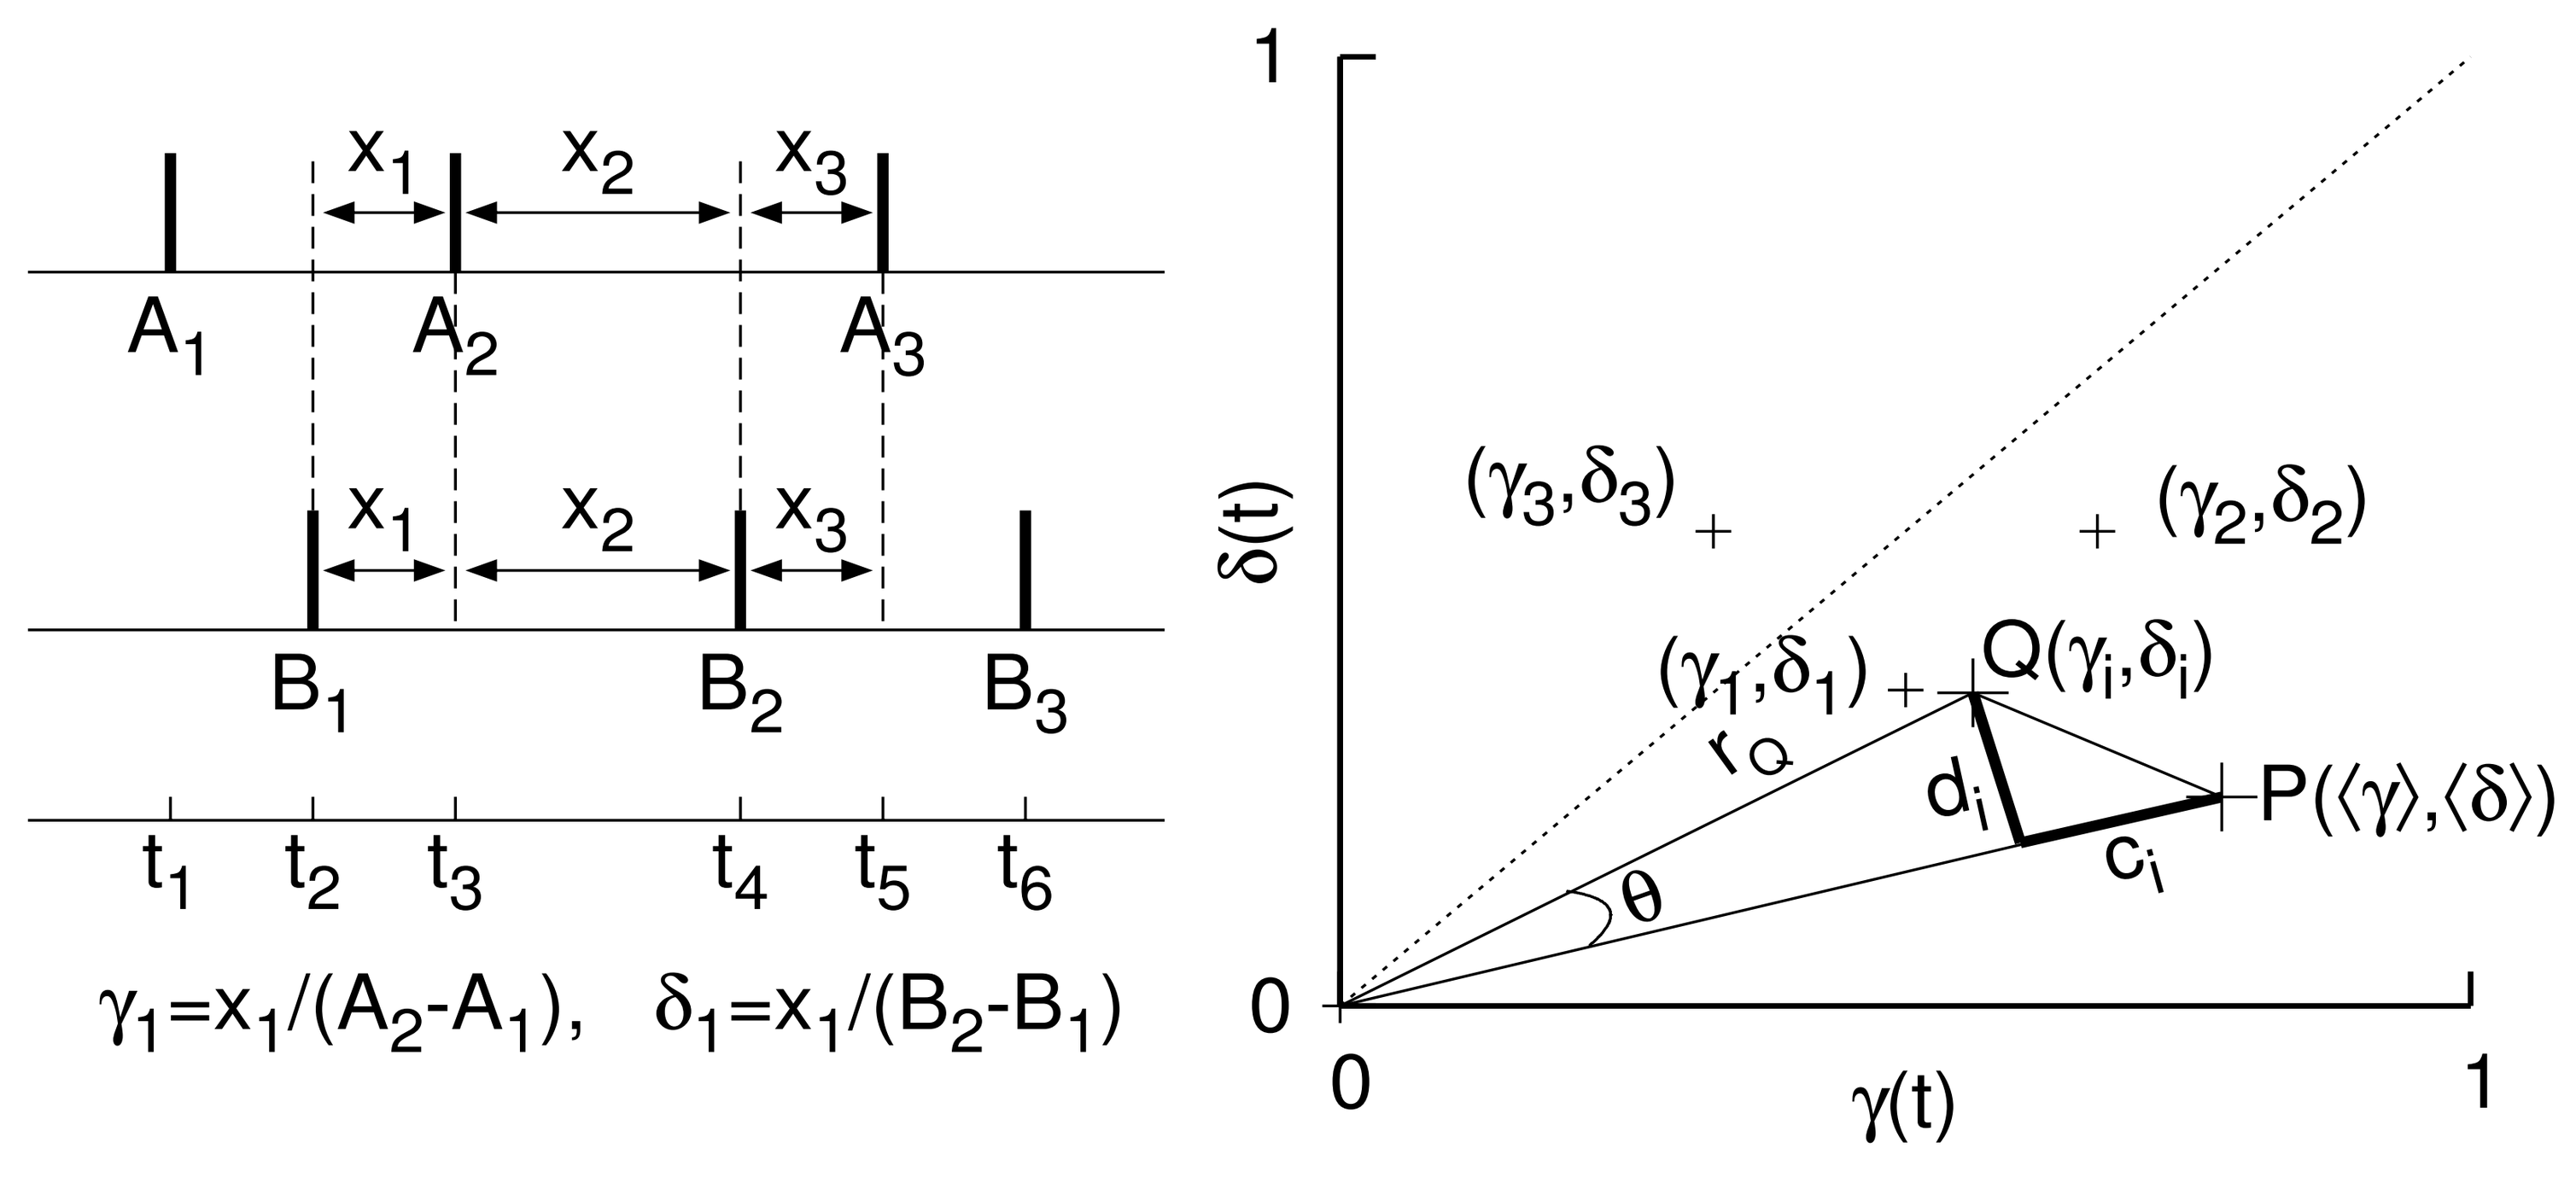
x_1 x_2 x_3
A_1 A_2 A_3
B_1 B_2 B_3
t_1 t_2 t_3 t_4 t_5 t_6
$\gamma_1=x_1/(A_2-A_1)$, $\delta_1=x_1/(B_2-B_1)$
(γ_3,δ_3) (γ_2,δ_2) (γ_1,δ_1) $Q(\gamma_i,\delta_i)$ $P(\langle\gamma\rangle,\langle\delta\rangle)$
r_Q d_i c_i θ
$\delta(t)$
$\gamma(t)$

- $A_1, A_2, A_3, \ldots, A_{N_A}$ are events of spike train A.
- $B_1, B_2, B_3, \ldots, B_{N_B}$ are events of spike train B.
- B-train is assumed to be time shifted by τ.
- Phase pairs (γ_i, δ_i) are constructed.
- $P(\langle\gamma\rangle, \langle\delta\rangle)$ is the mean of all phase points.
- OP is the mean phase curve, pairs locked in $\langle\gamma\rangle : \langle\delta\rangle$ ratio.
- The phase drift $c_i(\tau) = \frac{\gamma_i\langle\gamma\rangle+\delta_i\langle\delta\rangle}{\sqrt{\langle\gamma\rangle^2+\langle\delta\rangle^2}}$.
- Drift parameter, $C(\tau) = \frac{1}{N}\sum_{i=1}^{N} \mid c_i(\tau) \mid$.
- Phase function $\psi(\tau) = \frac{C(\tau)}{\sqrt{2}}$.

Phase function properties:

- For $\tau \to 0^+$ or 0^-,
 the phase pairs alternate between $(0,0)$ and $(1,1)$.
 Hence, $C(\tau \to 0^+) = C(\tau \to 0^-) = \frac{1}{\sqrt{2}}$, and ψ becomes 0.5.
- At $\tau = 0$, $\gamma_i = \delta_i = 1$. Hence $\psi(\tau = 0) = 0$.
- For an asynchronous state that has phase pairs
 homogeneously distributed on the line OP between 0 and 1,
 $C(\tau \to \infty) = \frac{1}{N}\sum_{i=1}^{N} \sqrt{2} \mid \langle\gamma\rangle - \gamma_i \mid = 0.25\sqrt{2}$.
 Hence $\psi(\tau \to \infty) = 0.25$.

Application to a Poisson train:

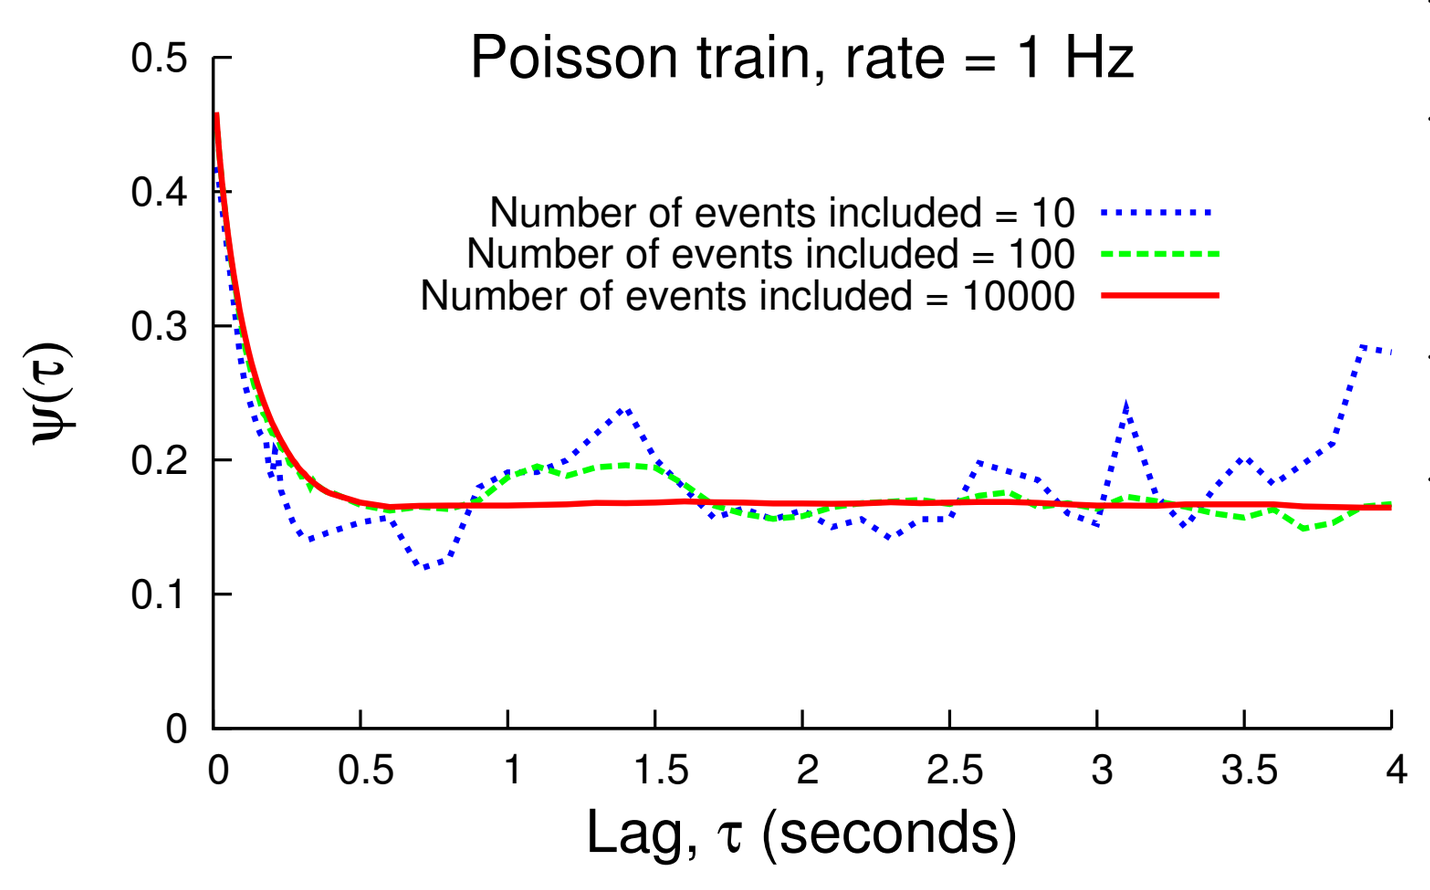
Poisson train, rate = 1 Hz
Number of events included = 10
Number of events included = 100
Number of events included = 10000
ψ(τ)
Lag, τ (seconds)

- ✶ 1 Hz Poisson spike train
- ✶ Autophase function for different length sequences
- ✶ For large samples, no oscillations.
- ✶ Decay near 0 lag is not a correlation time, but a decay time from the incoherent state near 0.

Autocorrelation vs. Autophase function:

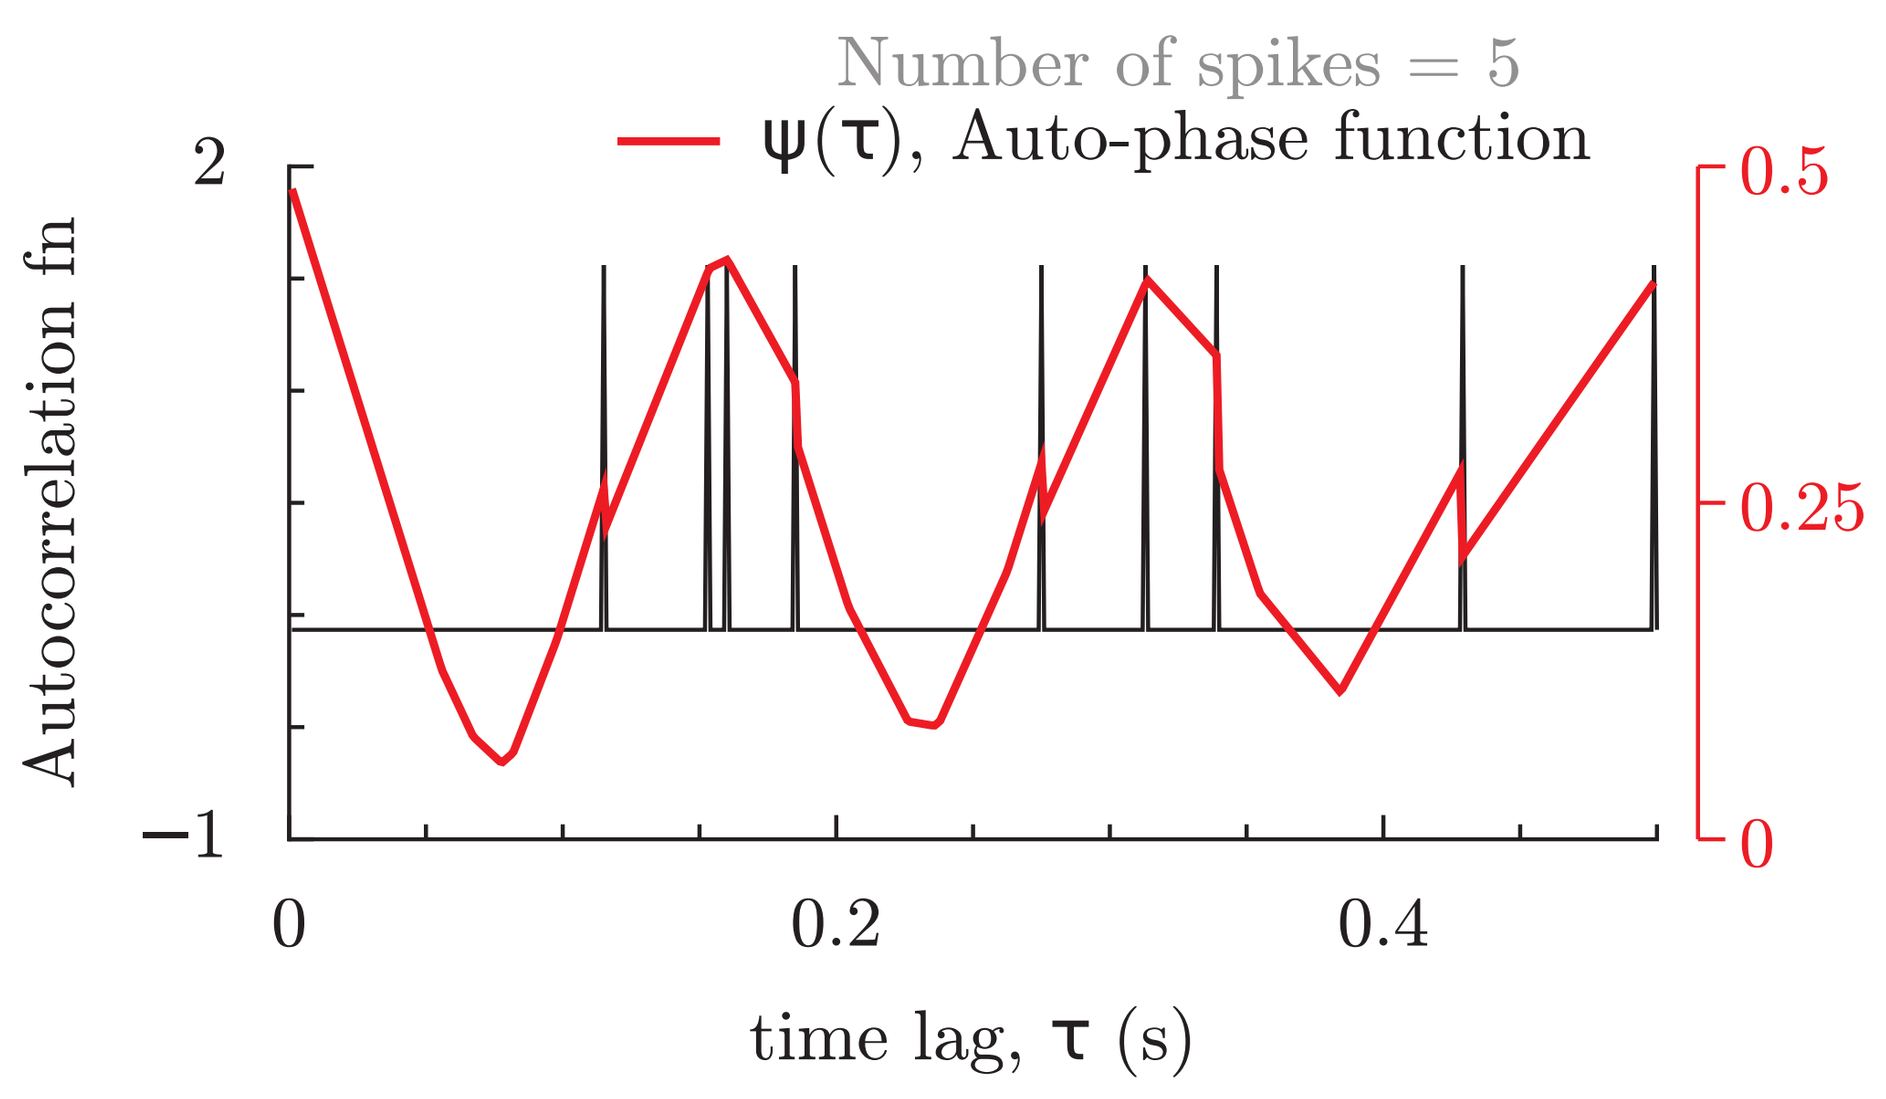
Number of spikes = 5
ψ(τ), Auto-phase function
Autocorrelation fn
time lag, τ (s)

✶ A brief dataset of 5 spike times

Table 1 (Time in sec)
0
0.18595
0.33955
0.4997
0.61495

- ✶ Phase function uses no smoothing.
- ✶ But shows superior profile than correlation function.

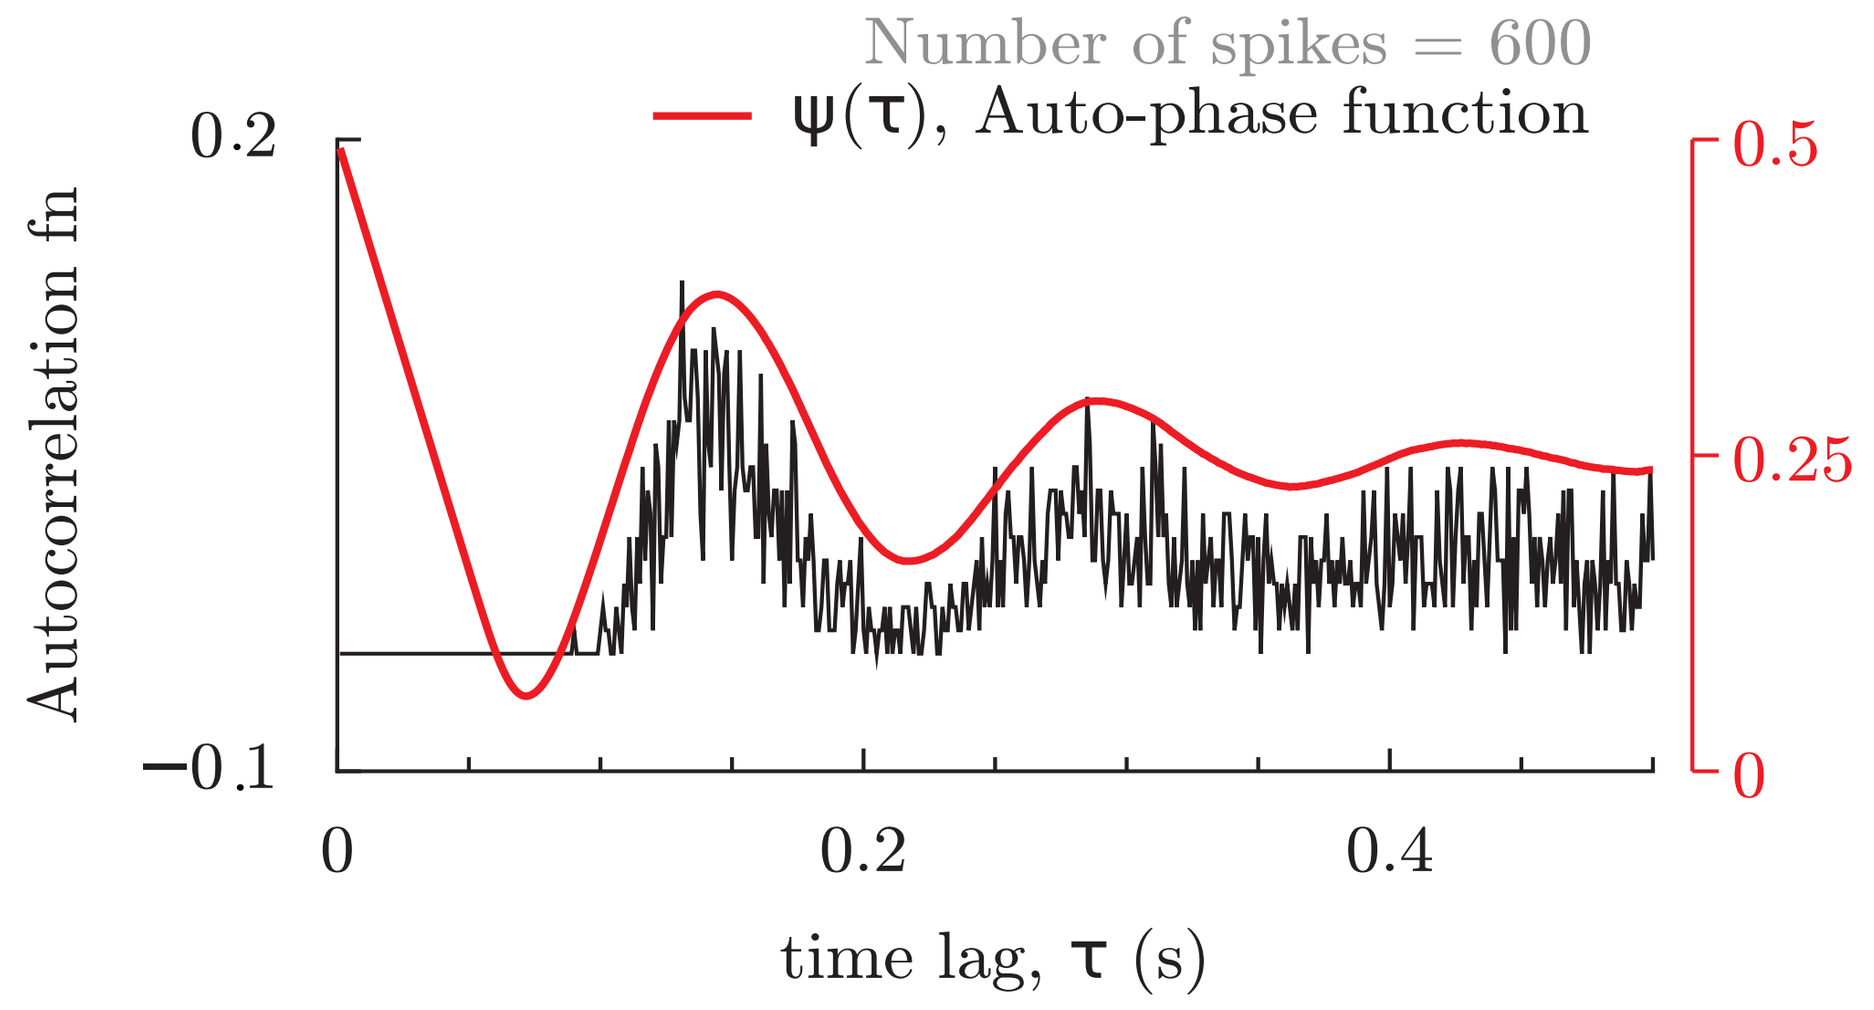
Number of spikes = 600
ψ(τ), Auto-phase function
Autocorrelation fn
time lag, τ (s)

Application to GPe recordings:

- ✶ Phase function for various length ISI sequences
- ✶ Comparison with autocorrelation function

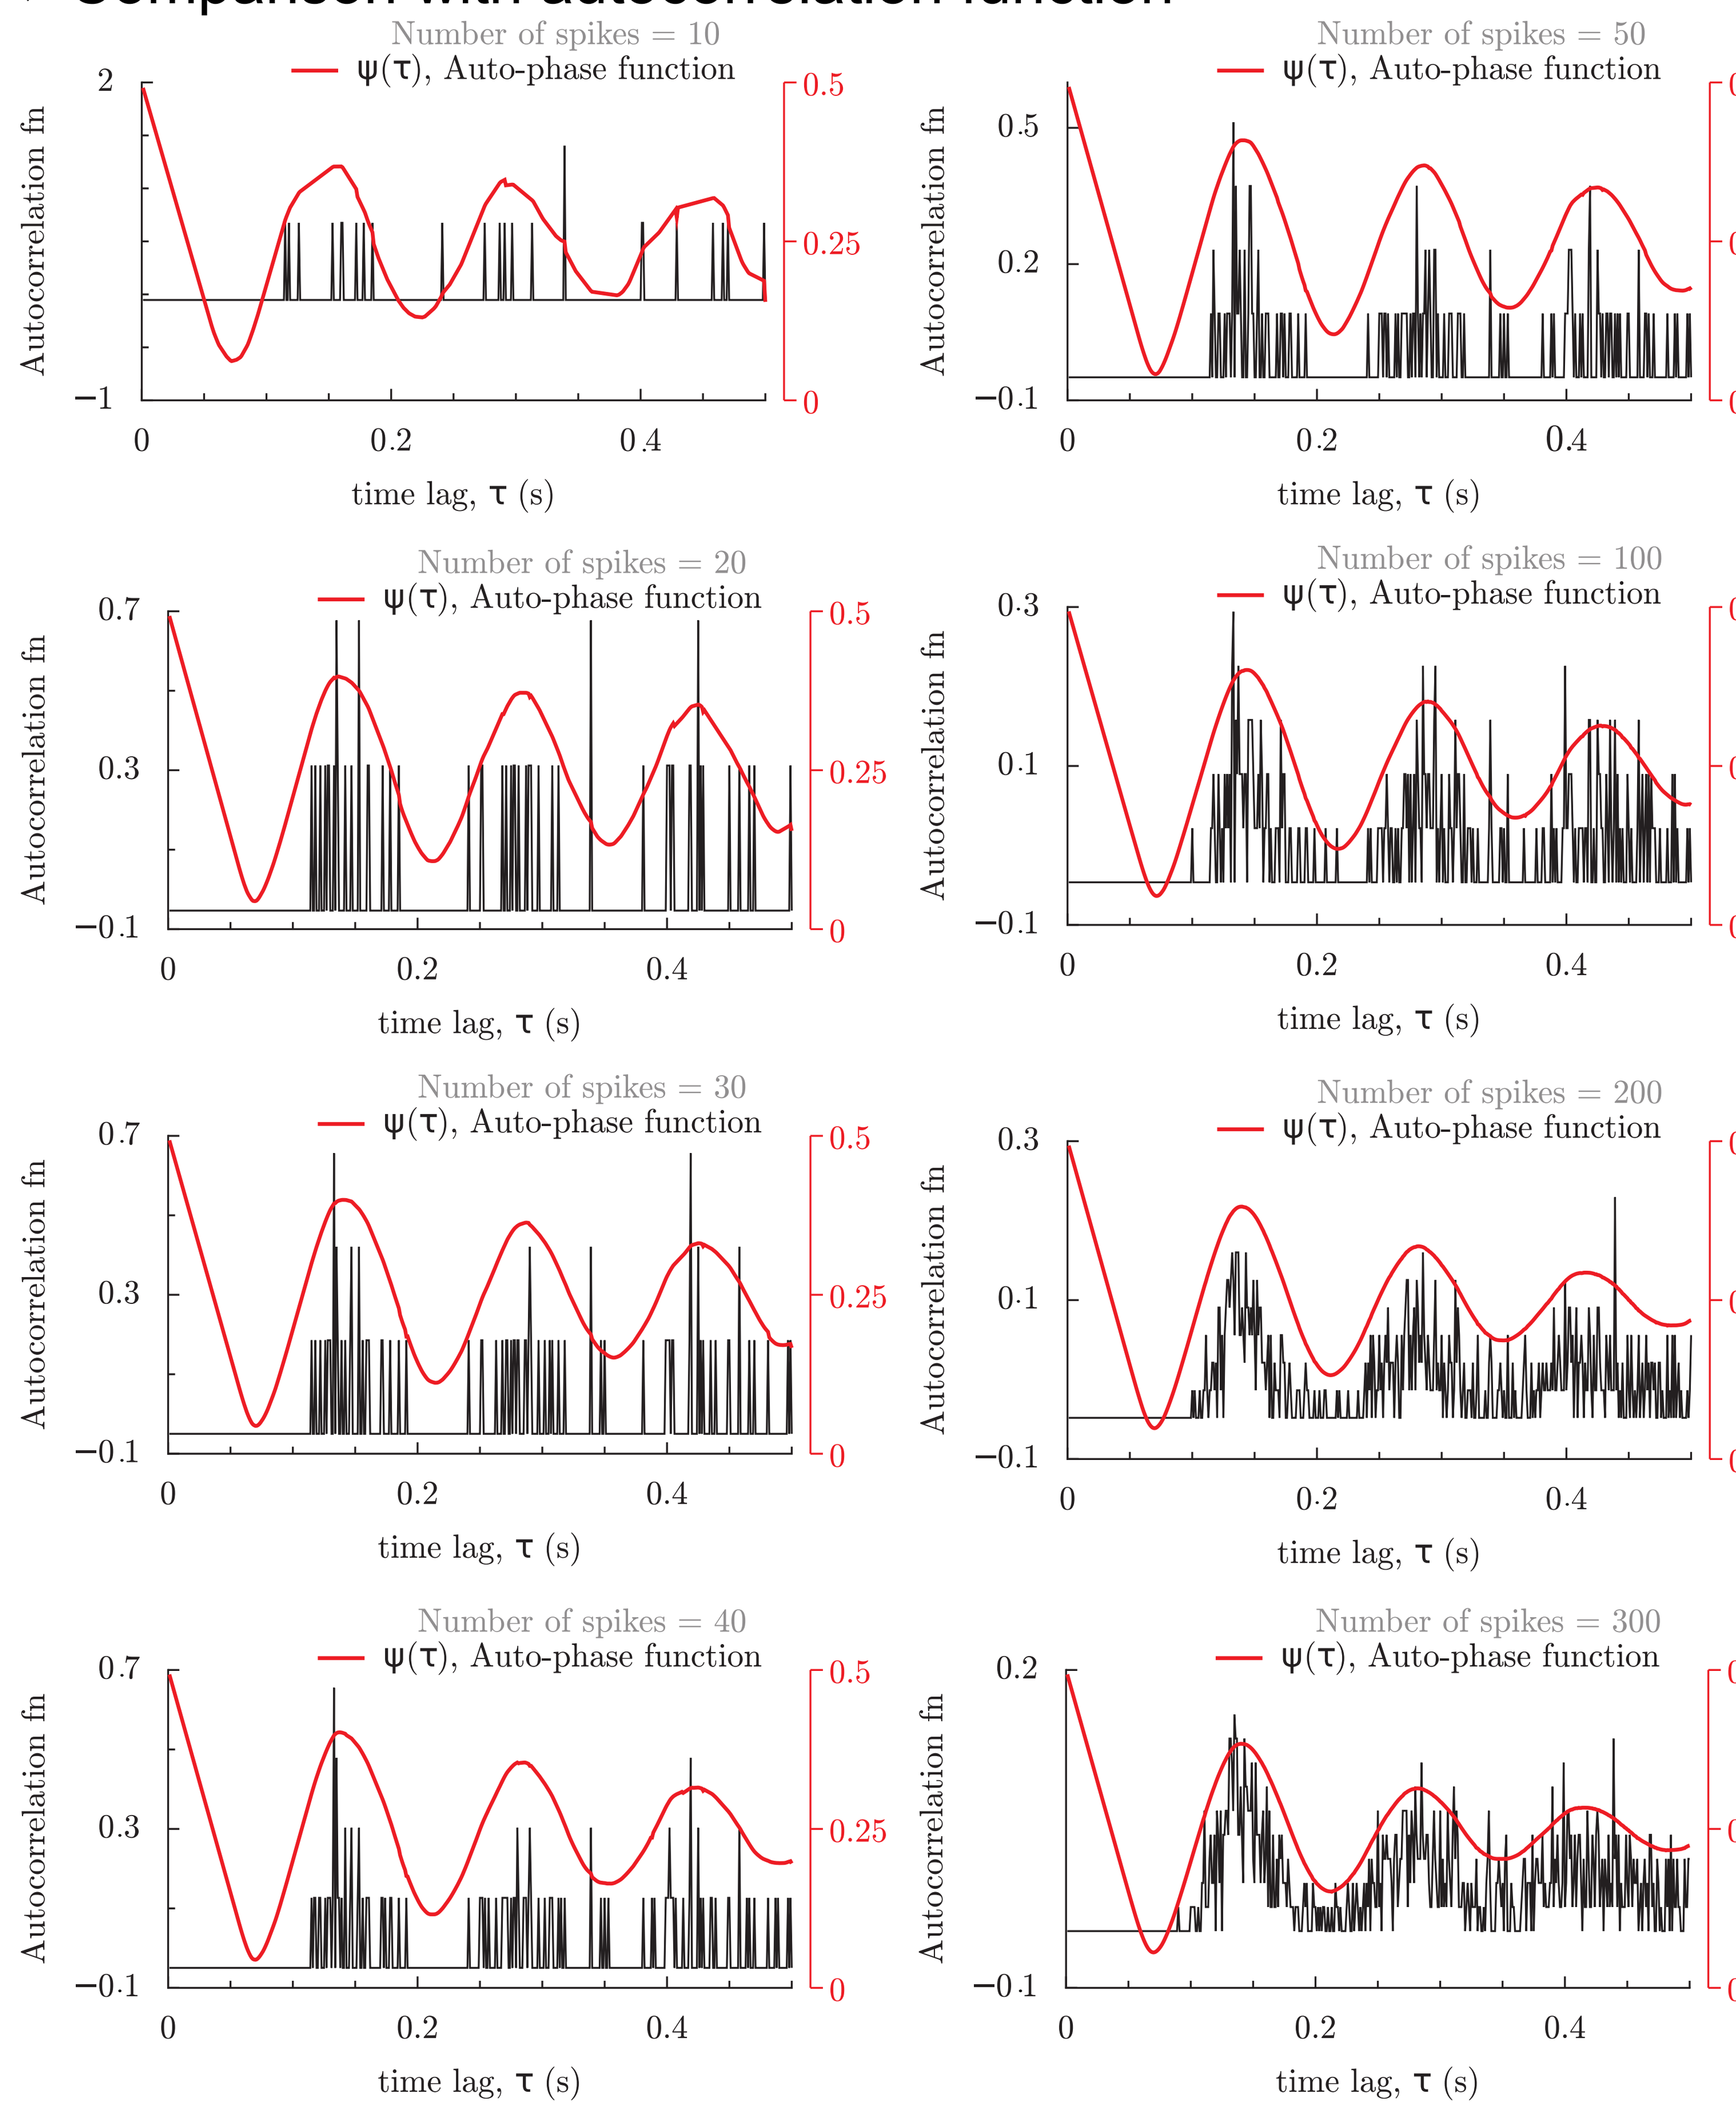
Number of spikes = 10
Number of spikes = 50
Number of spikes = 20
Number of spikes = 100
Number of spikes = 30
Number of spikes = 200
Number of spikes = 40
Number of spikes = 300
ψ(τ), Auto-phase function
Autocorrelation fn
time lag, τ (s)

Sliding window reveals periodicity:

✶ A sliding window of 5 ISIs used for computing phase function reveals strong local periodicity.

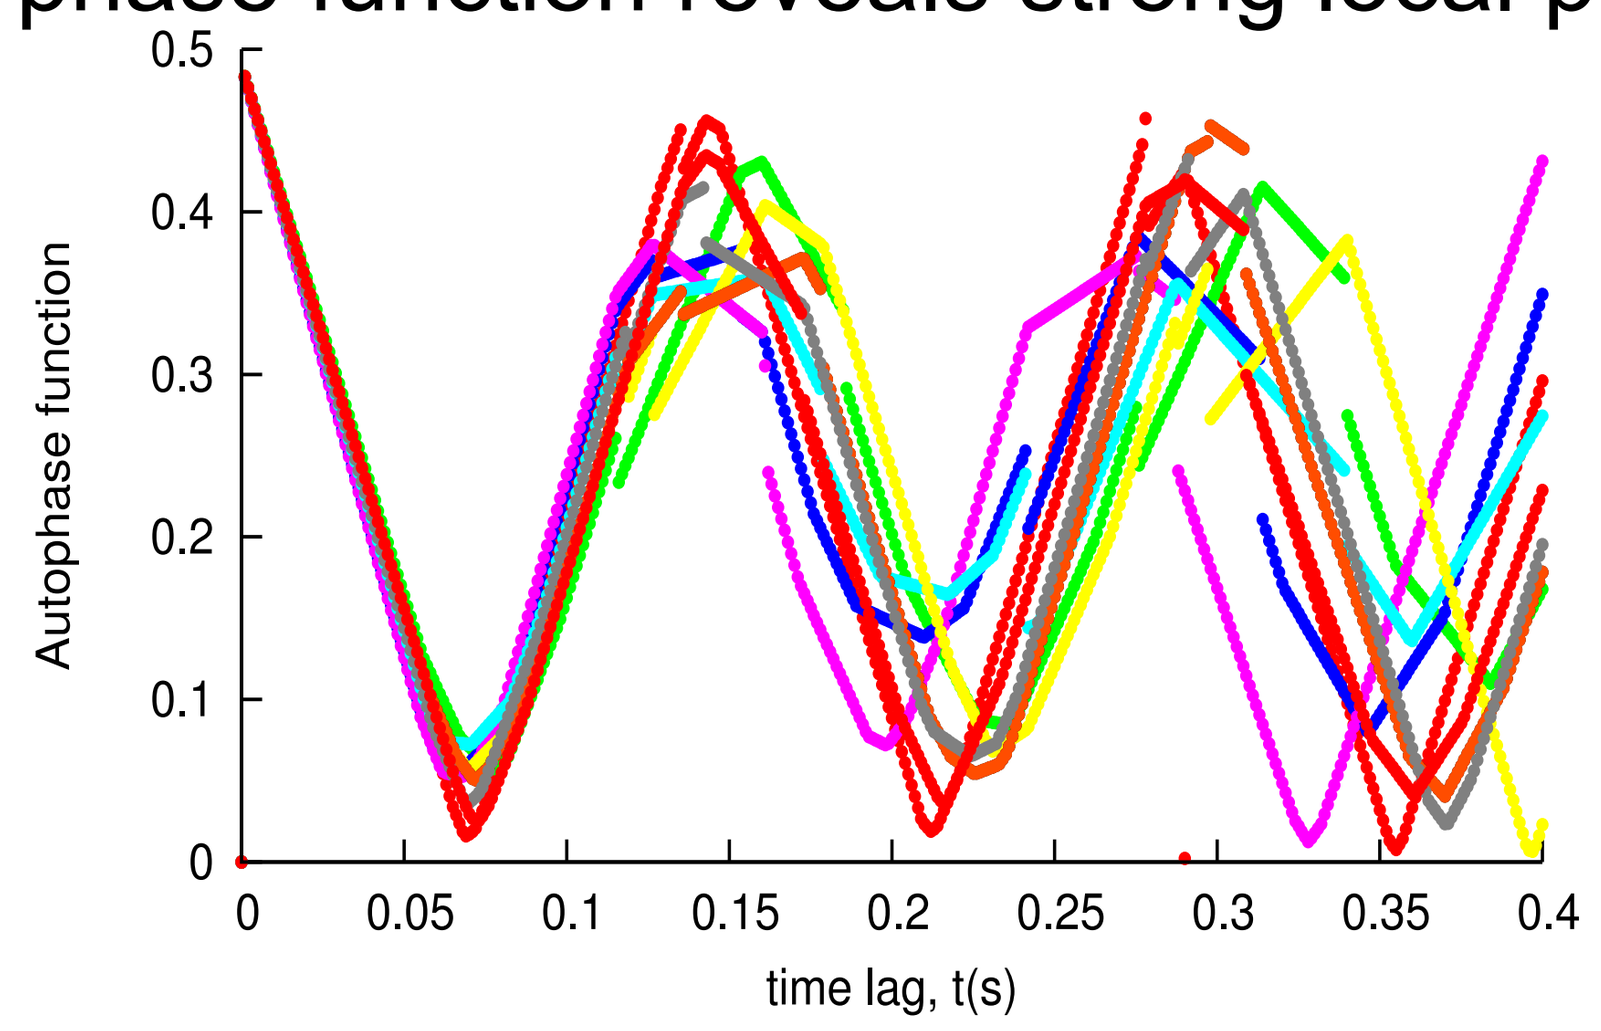
Autophase function
time lag, t(s)

ISI histogram:

✶ But the ISI density is indicative of the randomness of the ISIs.

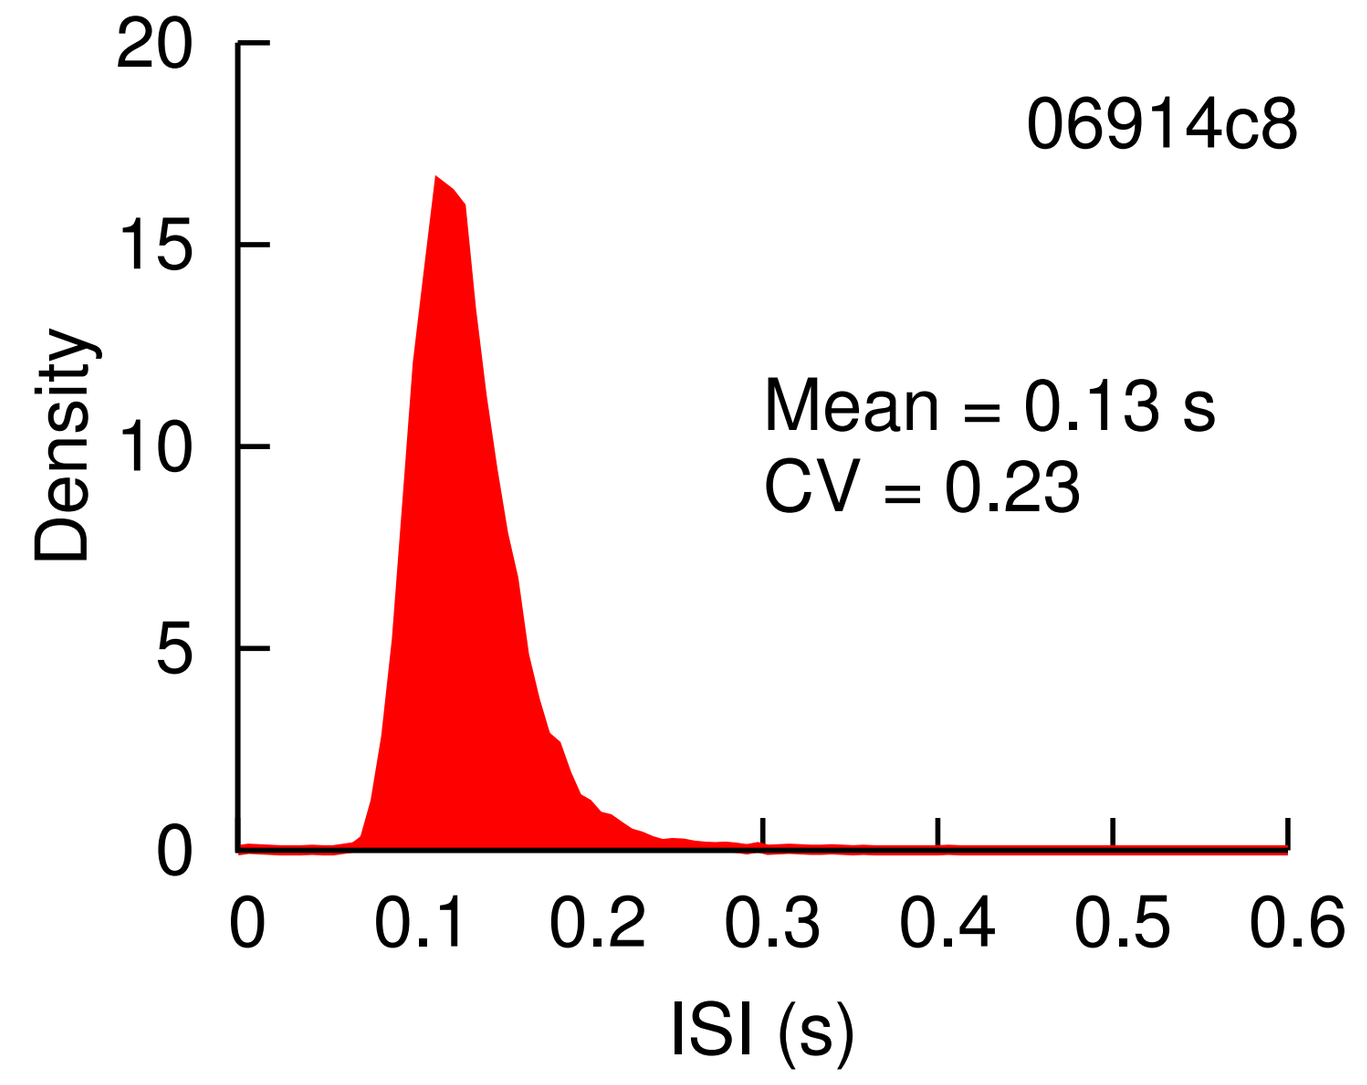
06914c8
Mean = 0.13 s
CV = 0.23
Density
ISI (s)

Nonstationarity and the nature of underlying noise

- ✶ The spike train recorded over one hour, displays nonstationarity.
- ✶ Mean firing rate for groups of consecutive ISIs without any transformation is shown below.
- ✶ The nonstationarity is partially but not totally removed by successive differencing of ISIs.
- ✶ It is unaffected by logarithmic transformation.

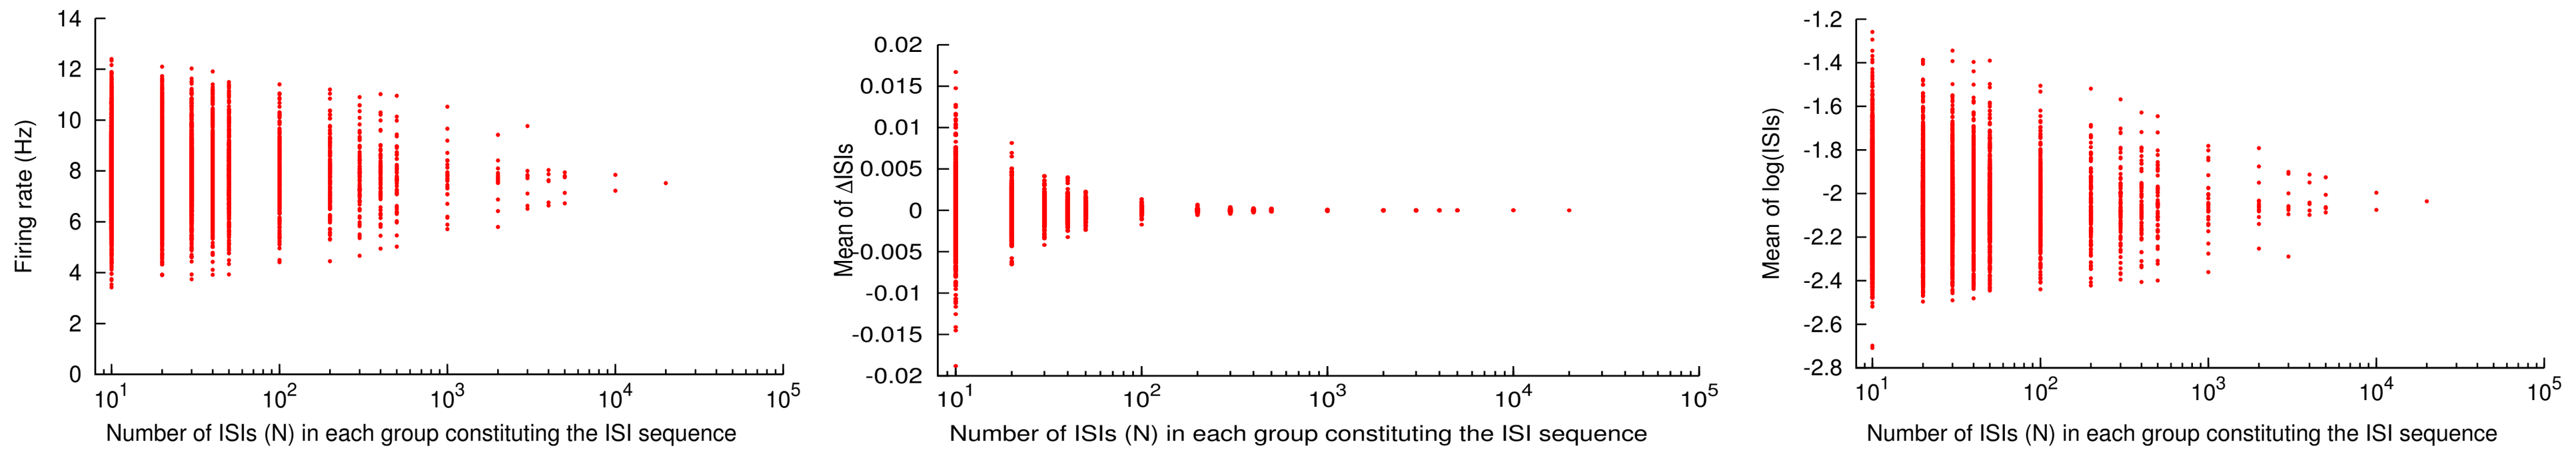
Firing rate (Hz)
Mean of ΔISIs
Mean of log(ISIs)
Number of ISIs (N) in each group constituting the ISI sequence

✶ Since the phase function analysis displays strong local periodicity, we subtract the local periodicity from the ISIs, and the resultant residuals are examined for normality.

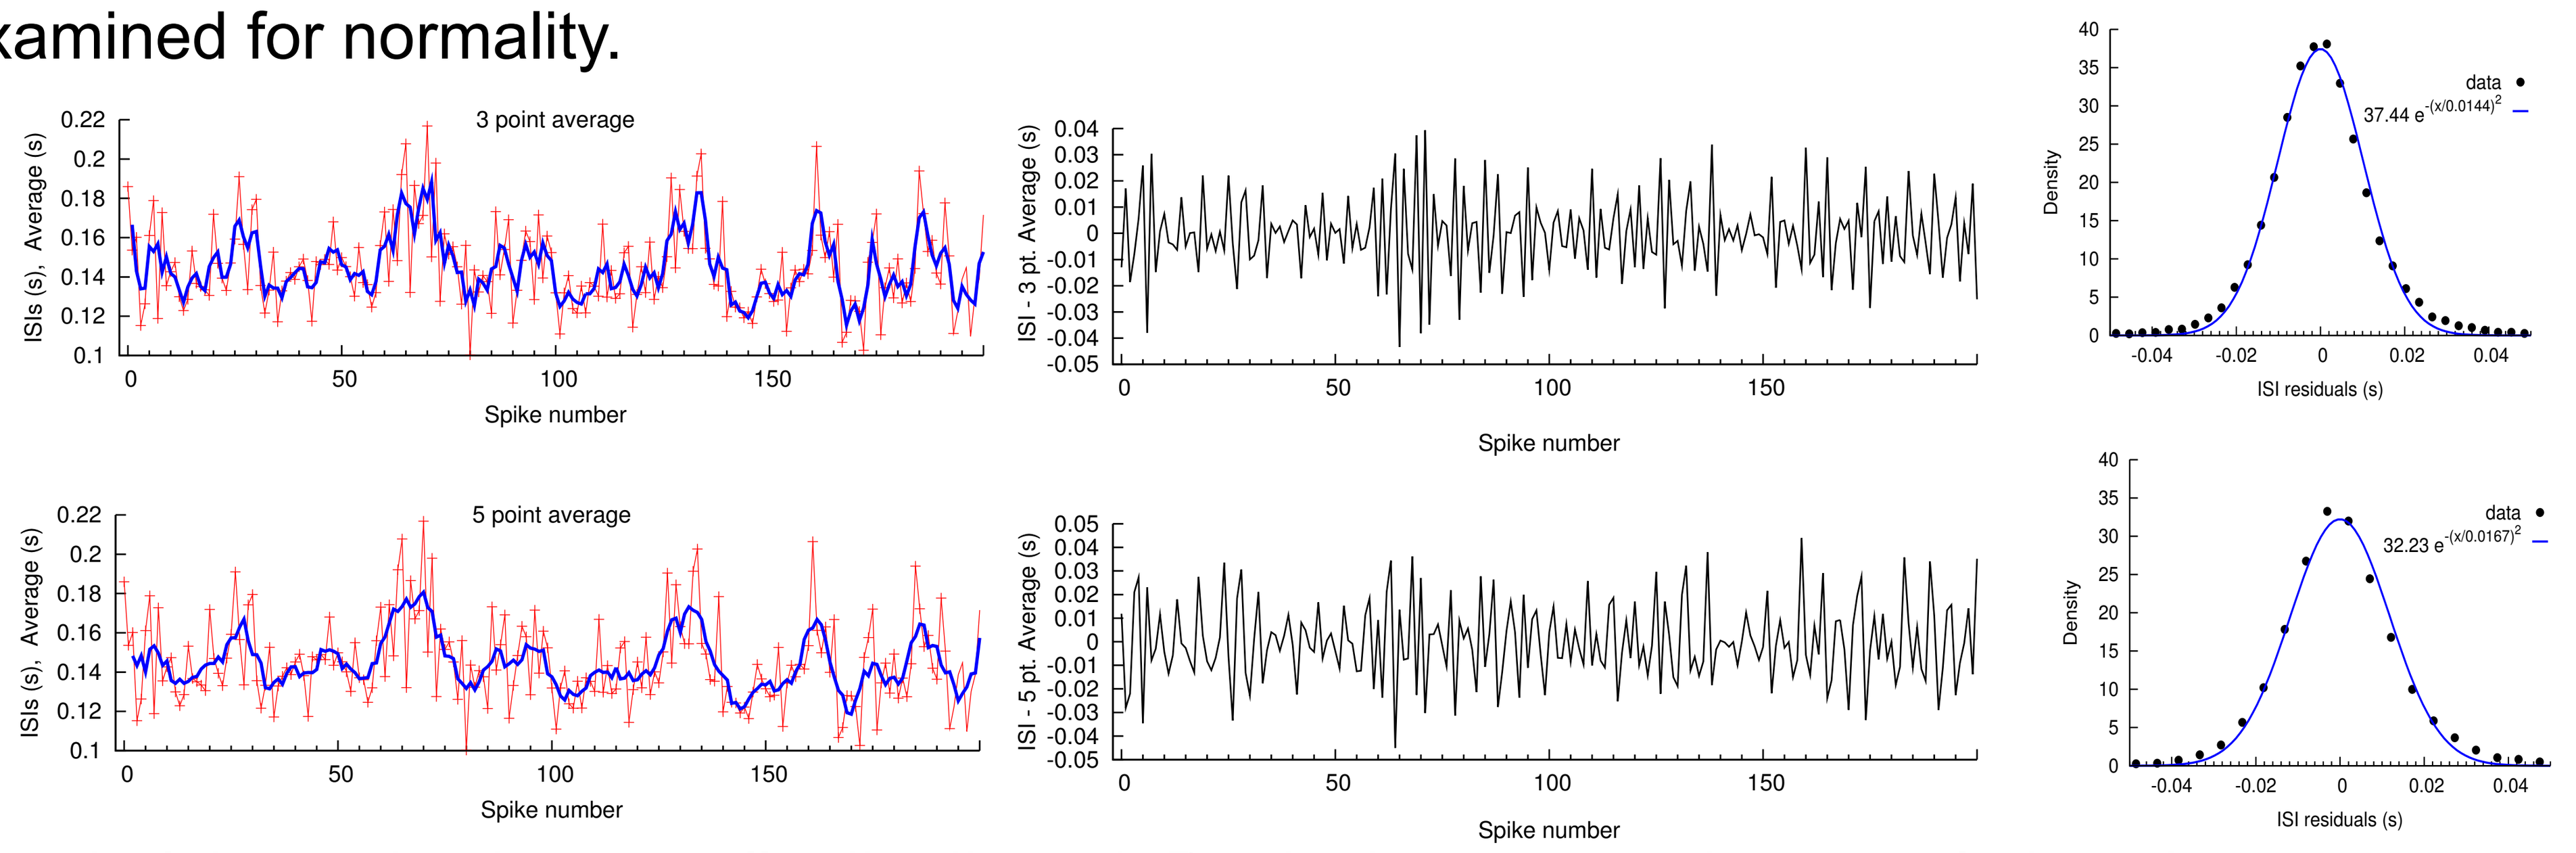
3 point average
5 point average
ISIs (s), Average (s)
ISI - 3 pt. Average (s)
ISI - 5 pt. Average (s)
Spike number
Density
data
ISI residuals (s)

✶ The underlying noise is normally distributed. But it shows negative correlations between neighoring terms.

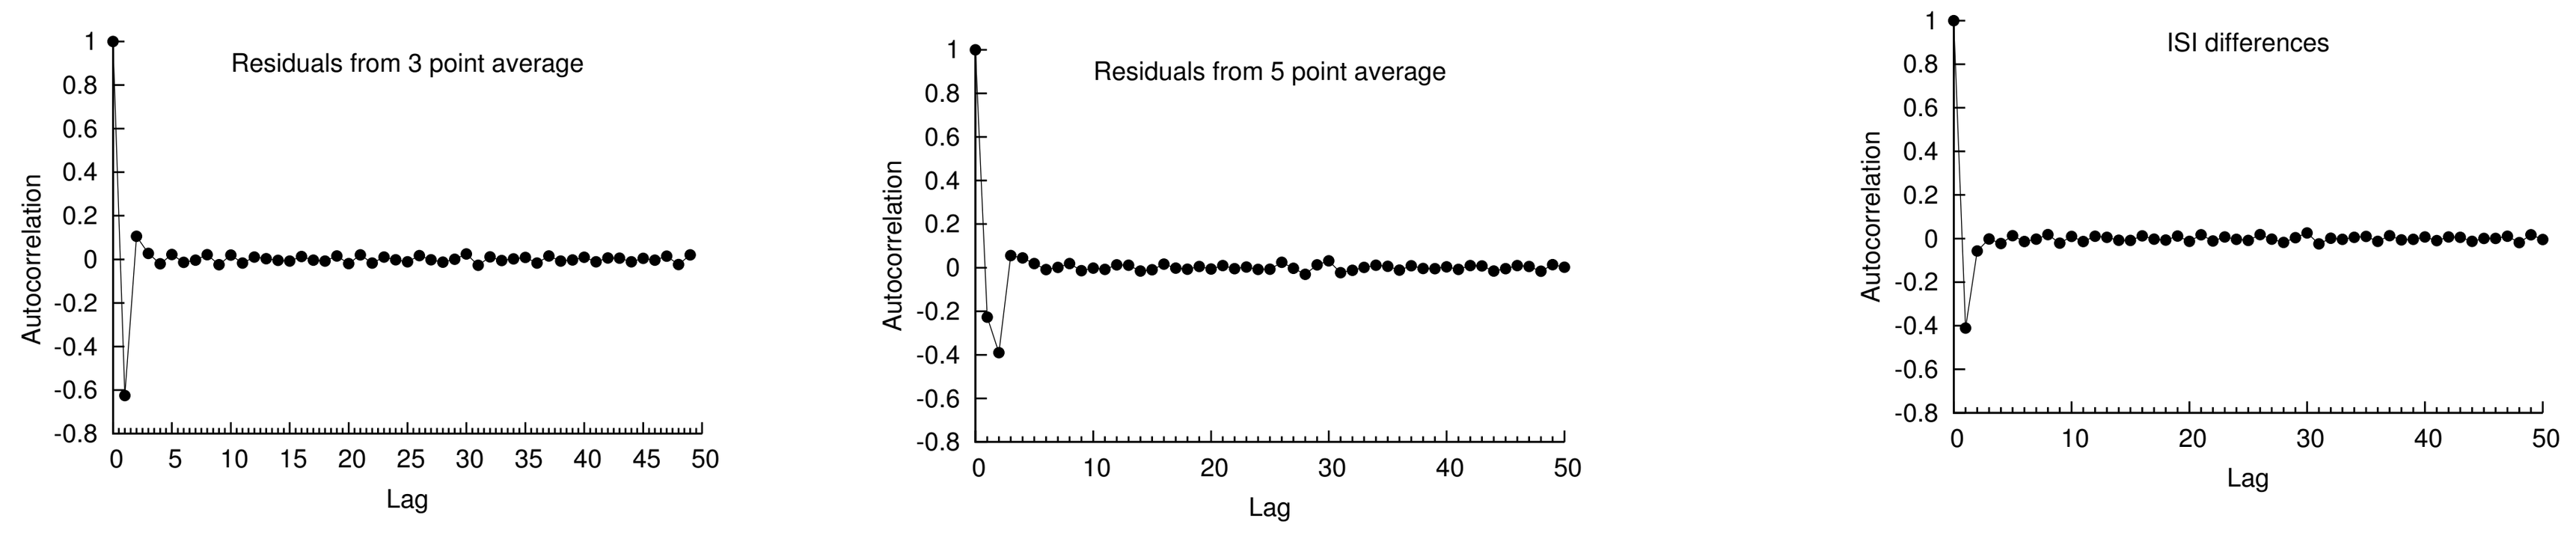
Residuals from 3 point average
Residuals from 5 point average
ISI differences
Autocorrelation
Lag

Conclusions:

- ✶ Phase function is demonstrated to perform better than correlation function when applied to discrete spike time sequences, particularly shorter ones.
- ✶ Globus Pallidus neurons are found to have strong periodicity at short time scales. The period however changes over time, but is affected by intrinsic noise component.
- ✶ When the periodicity is eliminated by subtracting the local mean ISI from each ISI, the resultant noise sequence is normal. The neighboring ISIs show negative correlations, but the sequence shows no long range correlations.