Synchrony-Asynchrony Transitions in Neuronal Networks

Ramana Dodla and Charles J. Wilson
Department of Biology, University of Texas at San Antonio, San Antonio, TX 78249

CNS 2008

Coupled Spiking Neuronal Networks

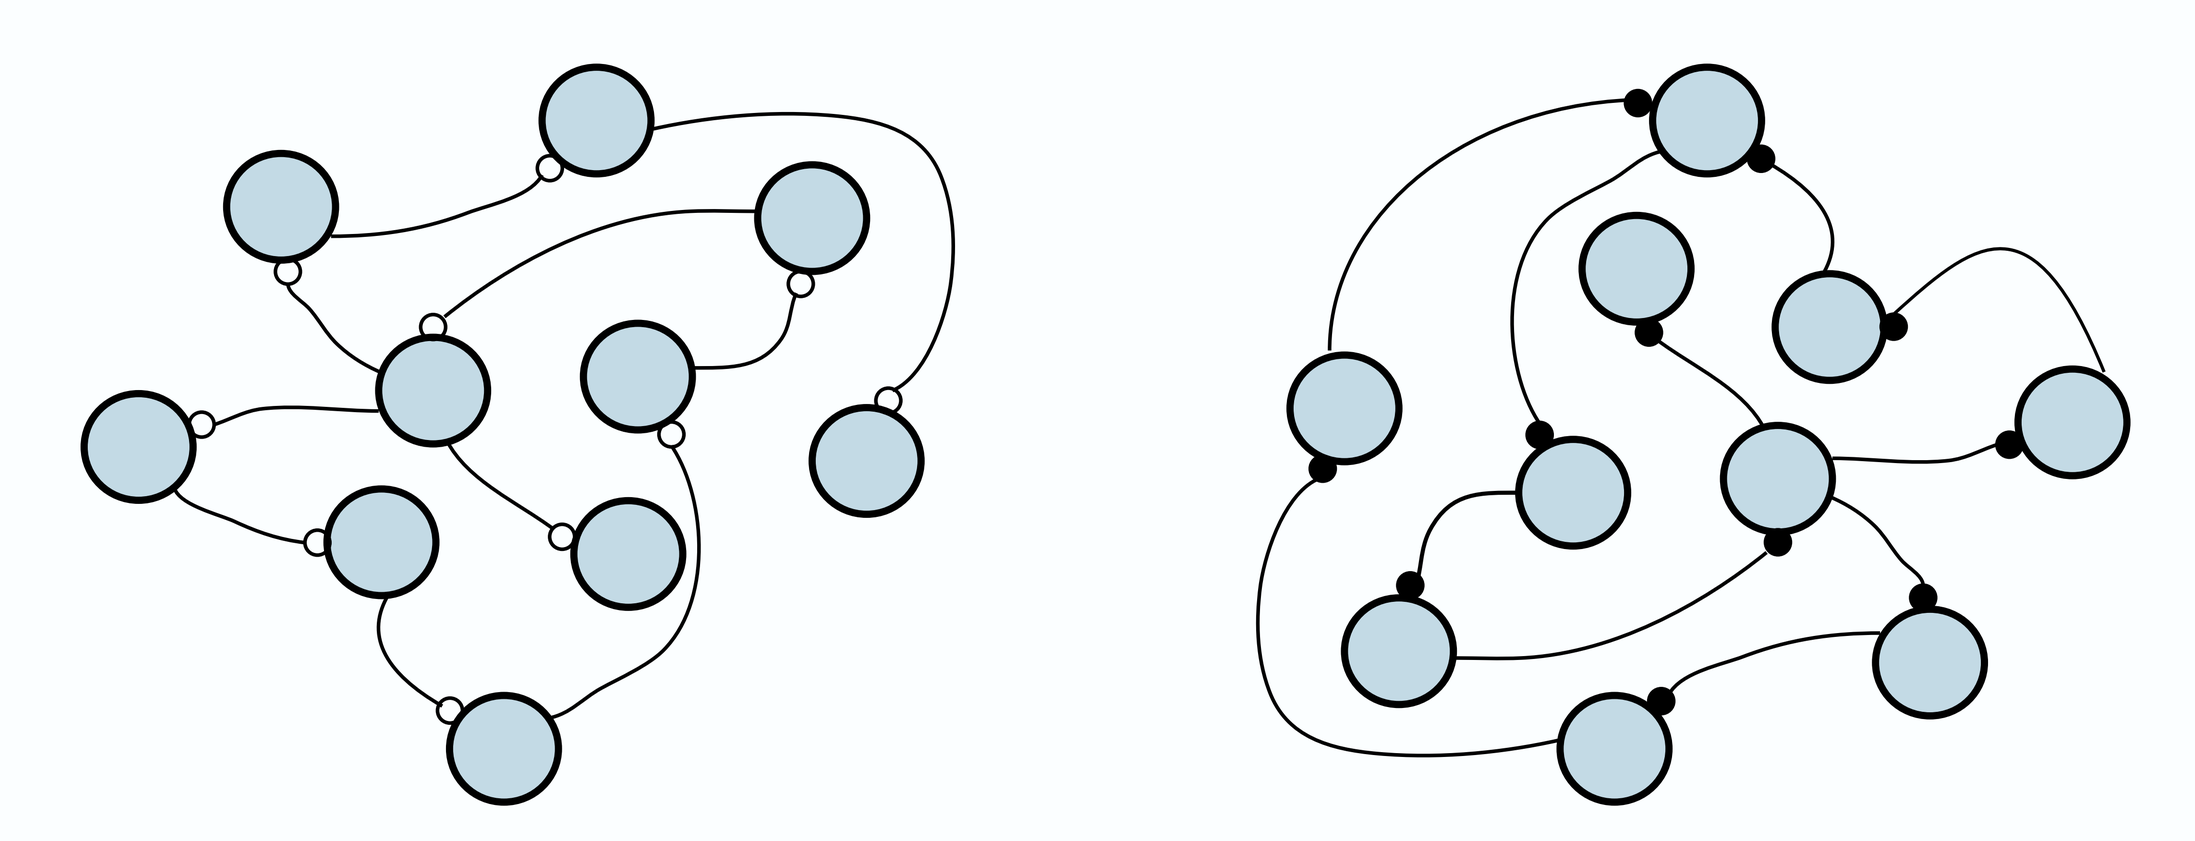

Subthalamic (excitatory) Hodgkin-Huxley (Inhibitory)

Motivation:

What are the conditions and nature of synchrony?
How does synaptic input modulate it?

Main Points:

1. Synchrony in EXC and INH Neurons
2. Synchrony-Asynchrony Transitions
3. Sparse Coupling, Breakdown of Synchrony
4. Loss of Synchrony Due to Synaptic Input

Model

$$\dot{V}_j = I_j - I_g^j - g\frac{1}{n}\sum_{k=1}^{N,k\neq j} M_{jk}S_k \times (V_j - E) - I_{syn}$$

j=1,.., N

Both excitatory (using STN neuron model) and inhibitory (using modified HH model) networks are considered.
M is the connection matrix.
Isyn is random shared input.

STN model:Terman et al. 2002

$$\dot{V} = I - I_{Na} - I_K - I_L - I_T - I_{AHP} - I_{Ca} - I_{syn},$$

$$\frac{dx}{dt} = \frac{x_\infty(V) - x}{\tau_x(V)},\ x = n, h, r,$$

$$\frac{d[Ca]}{dt} = 3.75\times10^{-5}(-I_{Ca} - I_T - 22.5[Ca]).$$

where

$$I_{Na} = G_{Na}m_\infty^3(V)h(V - E_{Na})$$

$$I_K = G_K n^4(V - E_K)$$

$$I_L = G_L(V - E_L)$$

$$I_T = G_T a_\infty^3(V)b_\infty^2(r)(V - E_{Ca})$$

$$I_{Ca} = G_{Ca}s_\infty^2(V)(V - E_{Ca})$$

$$I_{AHP} = G_{AHP}(V - E_K)\frac{[Ca]}{[Ca]+k}$$

PRC

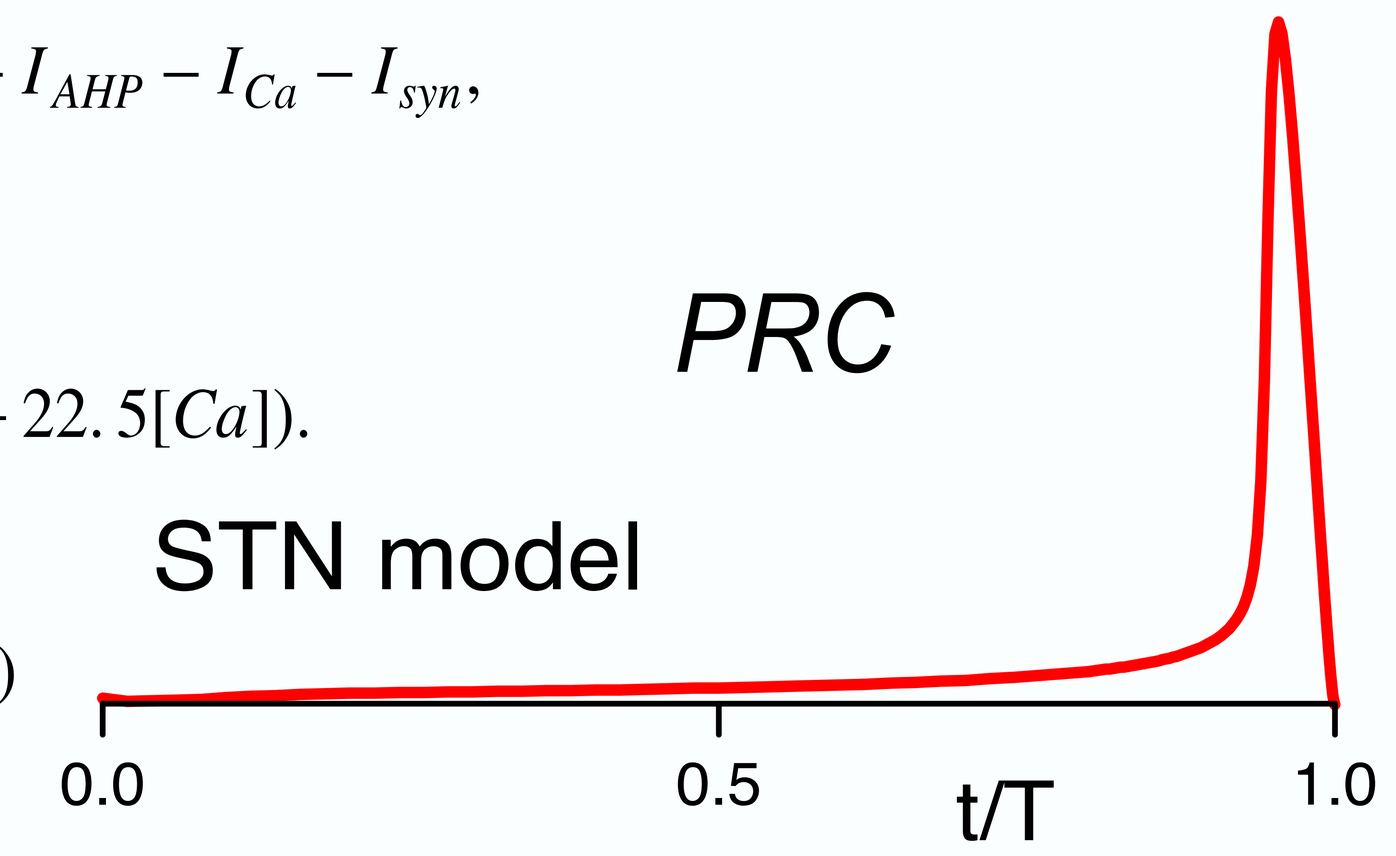
STN model
0.0
0.5
t/T
1.0

Type 1 like

HH model:
Hodgkin and Huxley 1952
with enhanced leakage reversal potential, so that pacemaking is triggered.

Type 2

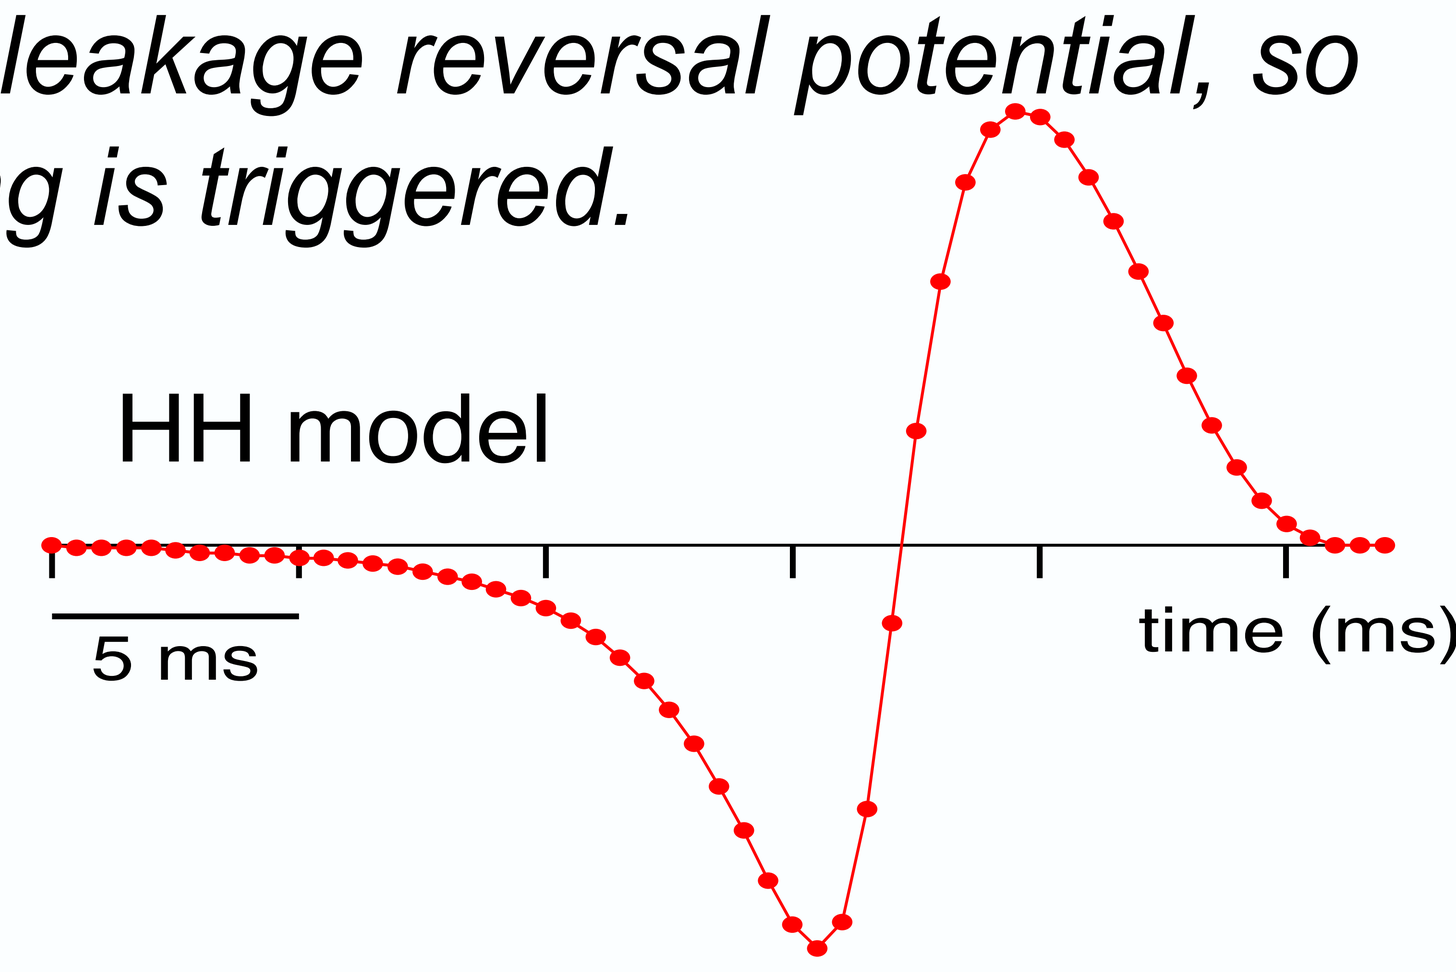
HH model
5 ms
time (ms)

Synchrony in 2 Coupled Excitatory STN Neurons

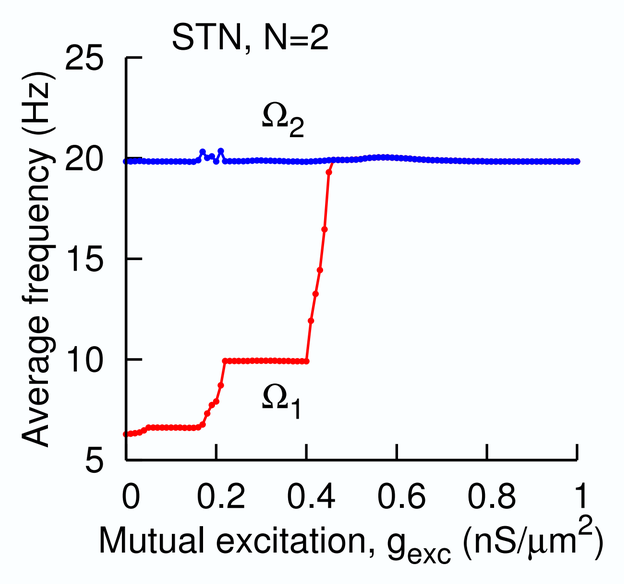
STN, N=2
Average frequency (Hz)
Mutual excitation, gexc (nS/μm2)

1:1 for synchrony can be achieved due to mutual excitatory coupling. For weak couplings, frequency lockings in m:n ratio can be found.

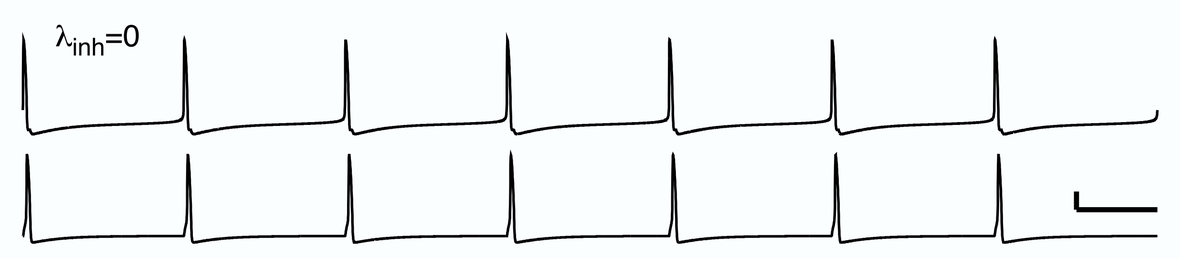

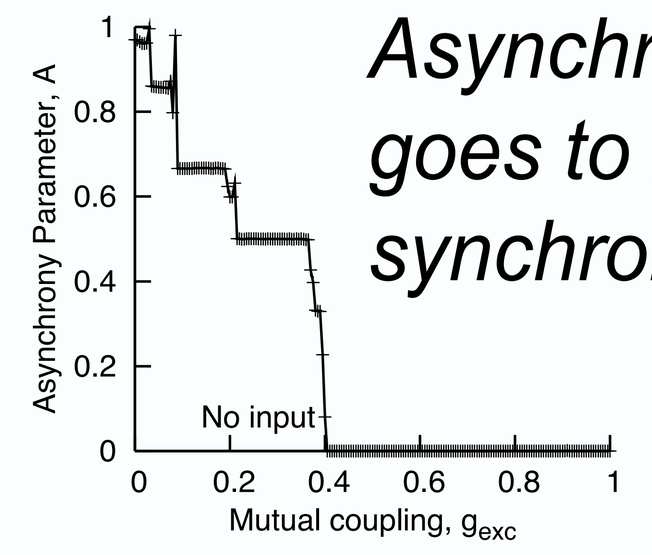
Asynchrony Parameter, A
No input
Mutual coupling, gexc

Asynchrony parameter goes to zero in the synchronized state.

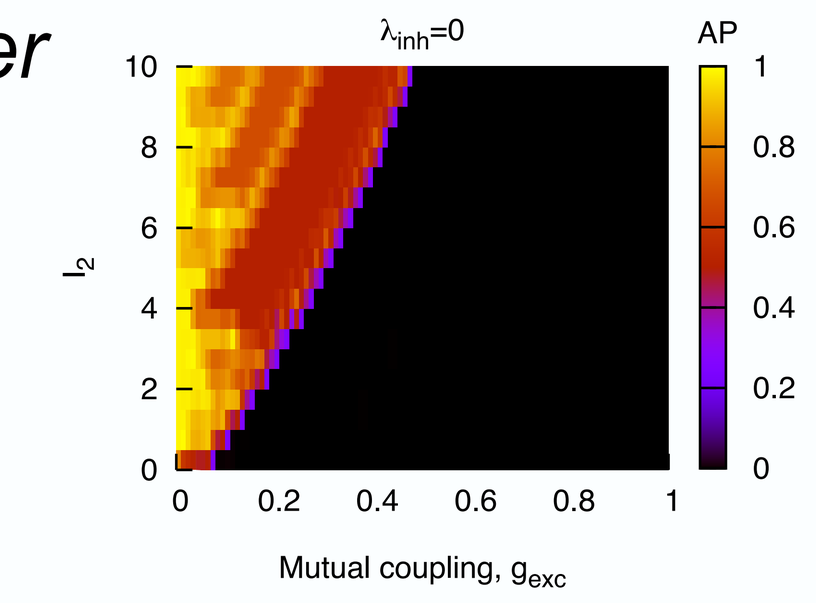
AP
Mutual coupling, gexc

Transition to synchrony takes place for all non-identical neurons.

Synchrony in Many Coupled Excitatory STN Neurons

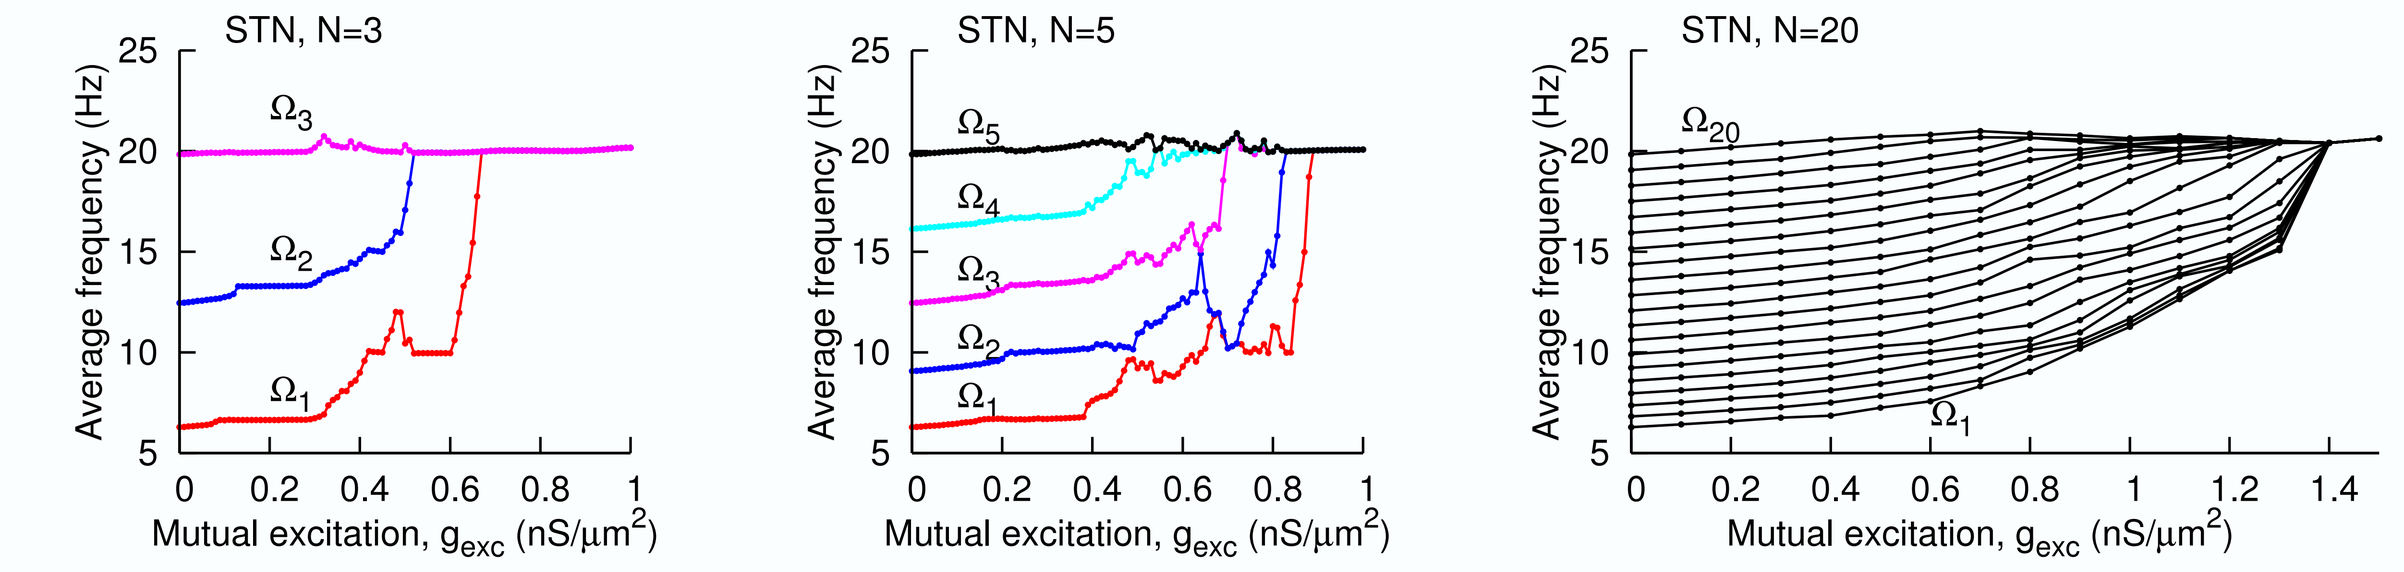
STN, N=3
STN, N=5
STN, N=20
Average frequency (Hz)
Mutual excitation, gexc (nS/μm2)

Synchrony in 2 or More Coupled Inhibitory HH Neurons

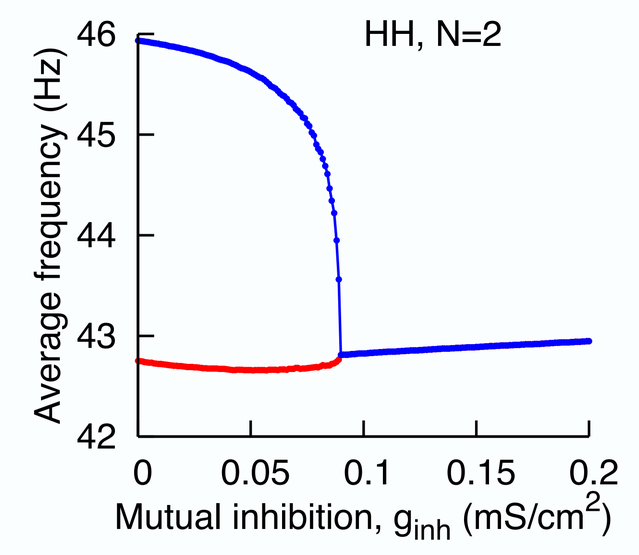
HH, N=2
Average frequency (Hz)
Mutual inhibition, ginh (mS/cm2)

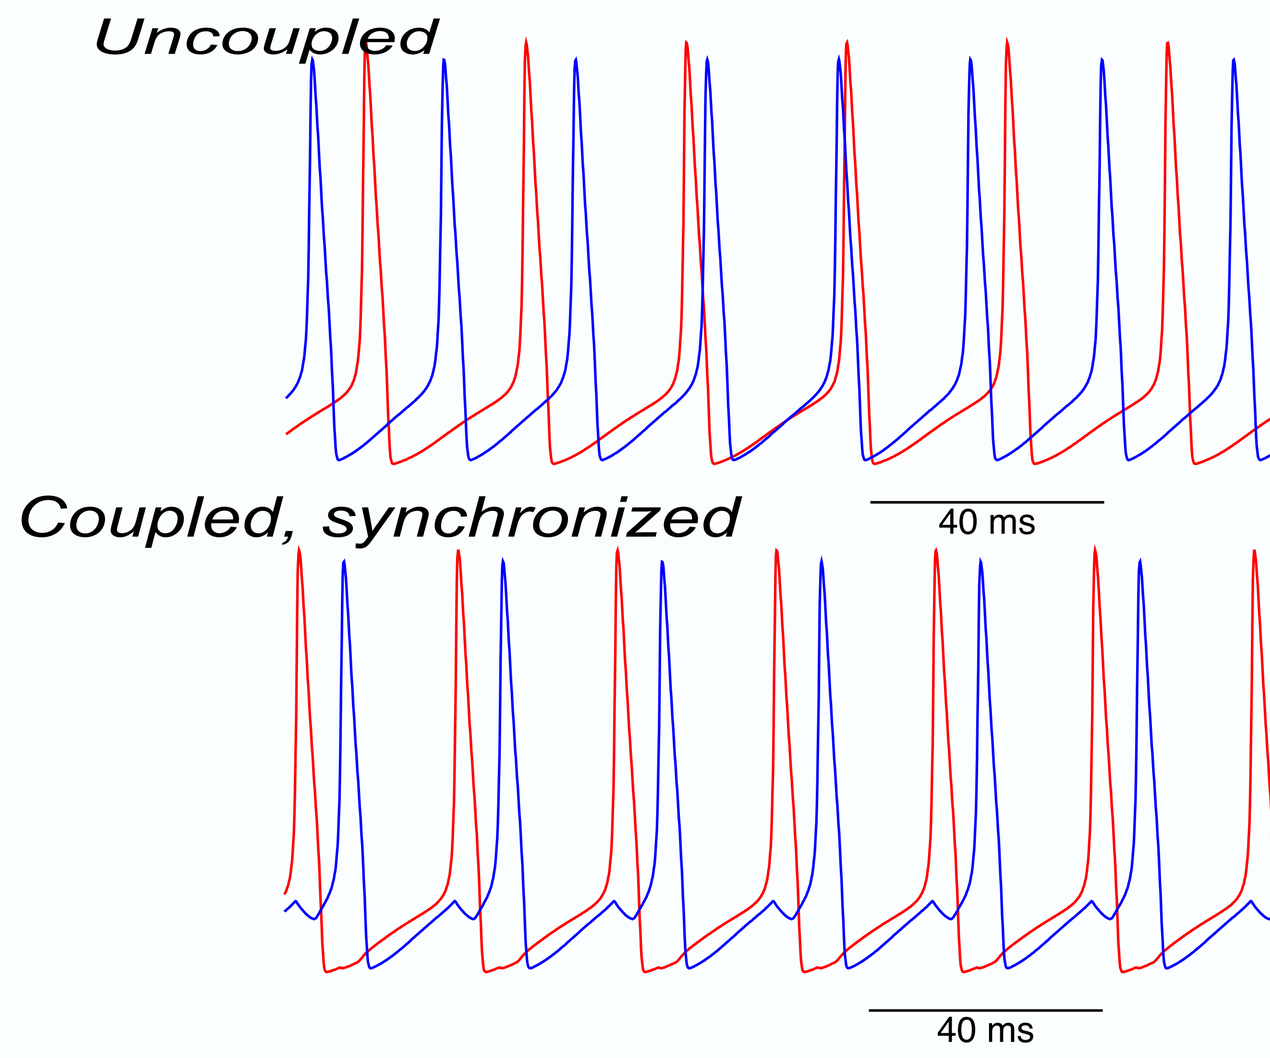
Uncoupled
40 ms
Coupled, synchronized
40 ms

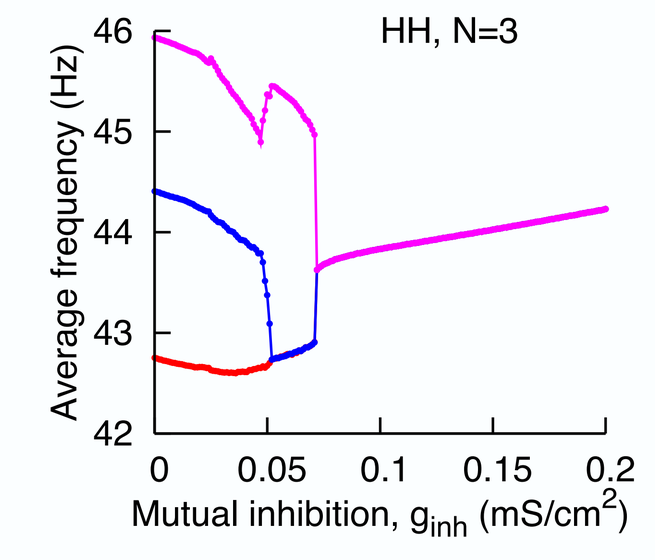
HH, N=3
Average frequency (Hz)
Mutual inhibition, ginh (mS/cm2)

1:1 for synchrony can also be achieved between two mutually inhibiting HH neurons. With global coupling (but no recurrent input), such synchrony occurs for weaker couplings.

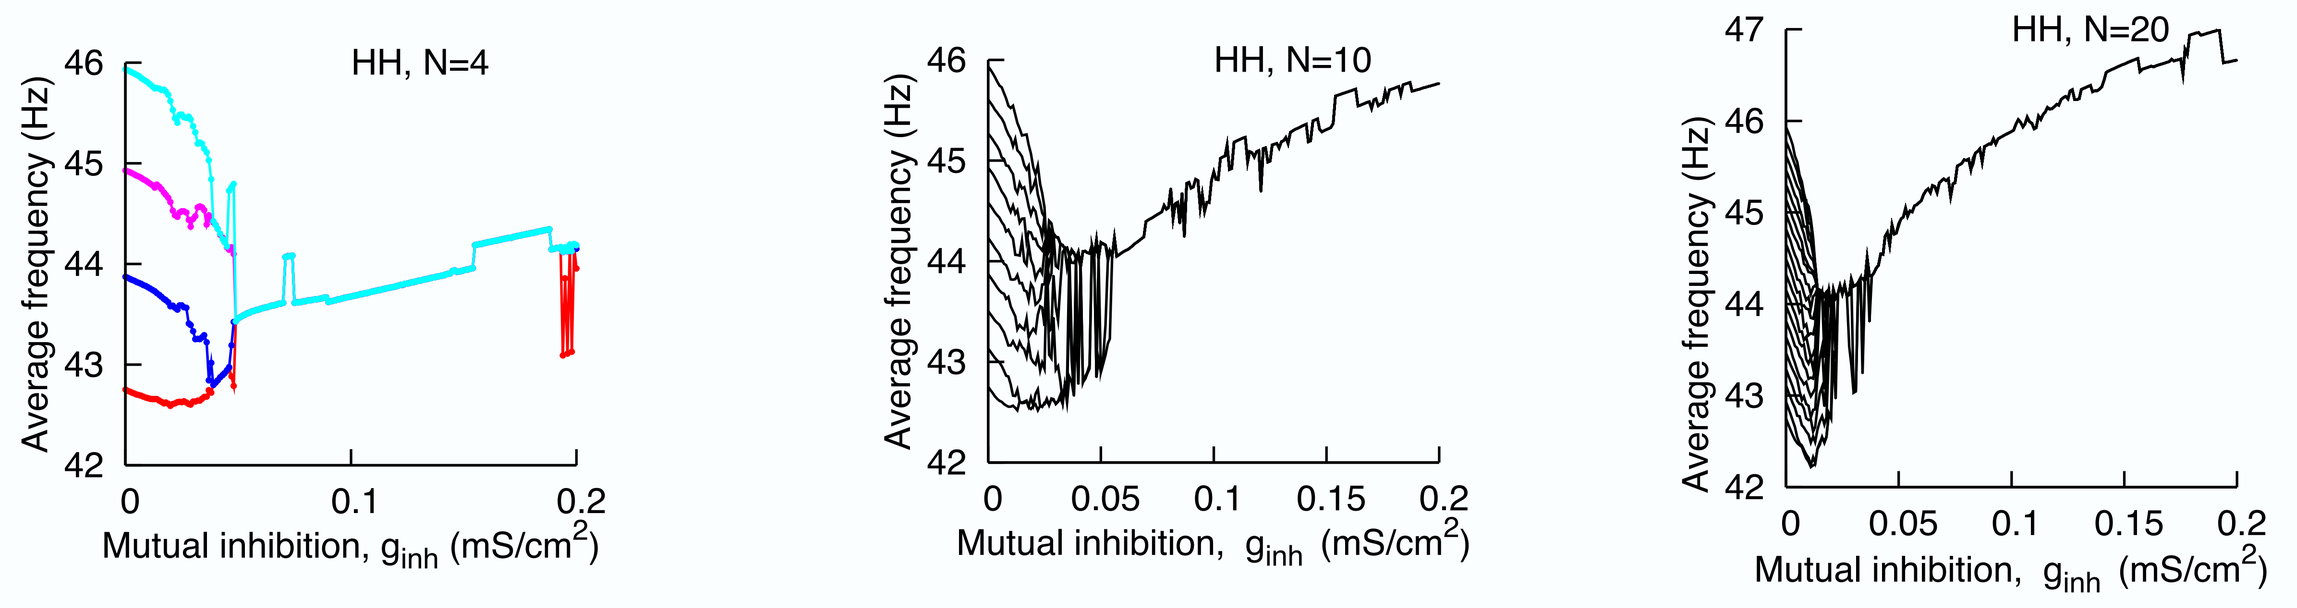
HH, N=4
HH, N=10
HH, N=20
Average frequency (Hz)
Mutual inhibition, ginh (mS/cm2)

Disorder Parameter, P

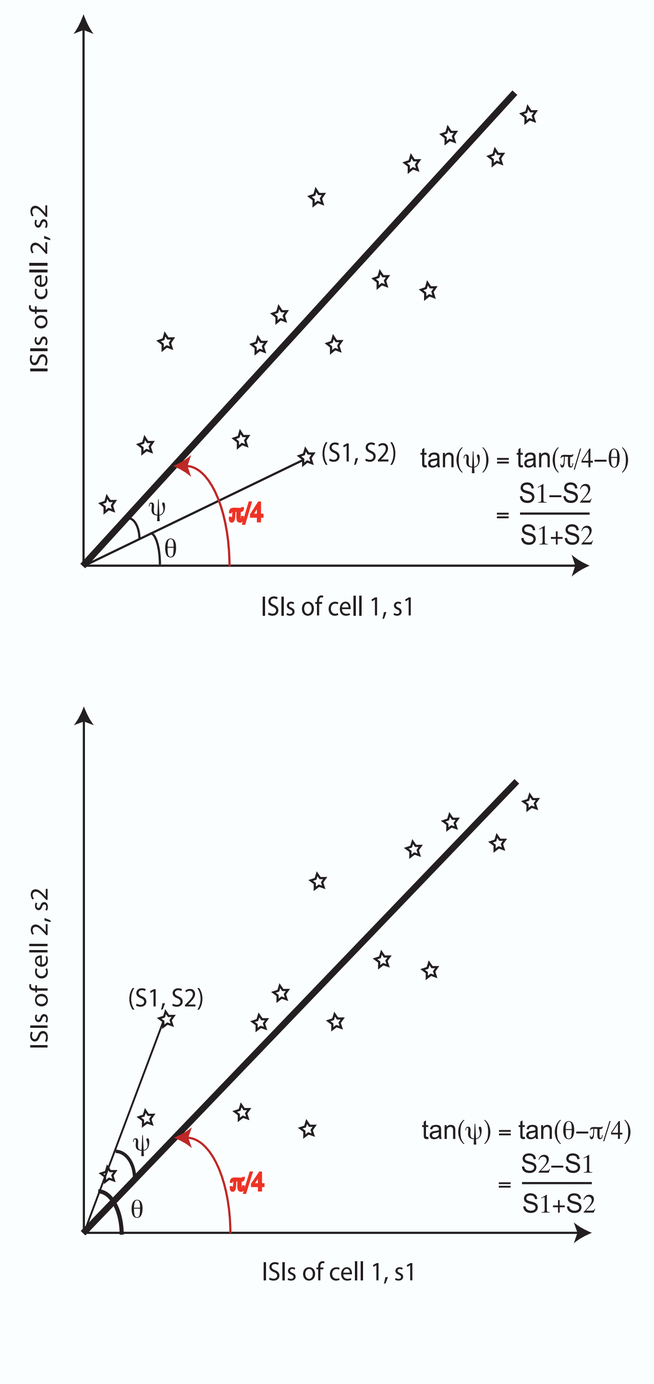
ISIs of cell 2, s2
ISIs of cell 1, s1
(S1, S2)
π/4
tan(ψ) = tan(π/4−θ) = (S1−S2)/(S1+S2)
tan(ψ) = tan(θ−π/4) = (S2−S1)/(S1+S2)

$$P = \frac{1}{N}\sum_{i=1}^{N}\tan\psi_i = \frac{1}{N}\sum_{i=1}^{N}\frac{|S^i{}_1 - S^i{}_2|}{S^i{}_1 + S^i{}_2}\,dt$$

$$\frac{1}{T}\int_0^T \tan\psi\ dt = \frac{1}{T}\int_0^T\frac{|S_1 - S_2|}{S_1 + S_2}\,dt$$

Geometrically, this definition is similar to "spike distance" measure of Kreuz et al. 2007.

Synchrony-Asynchrony Transitions

Dependence on N

1:1 synchrony is achieved in all-to-all coupled excitatory network (without self excitation) at different values of critical coupling strenghts. This value scales with the number of coupled neurons as, approximately, N^0.3.

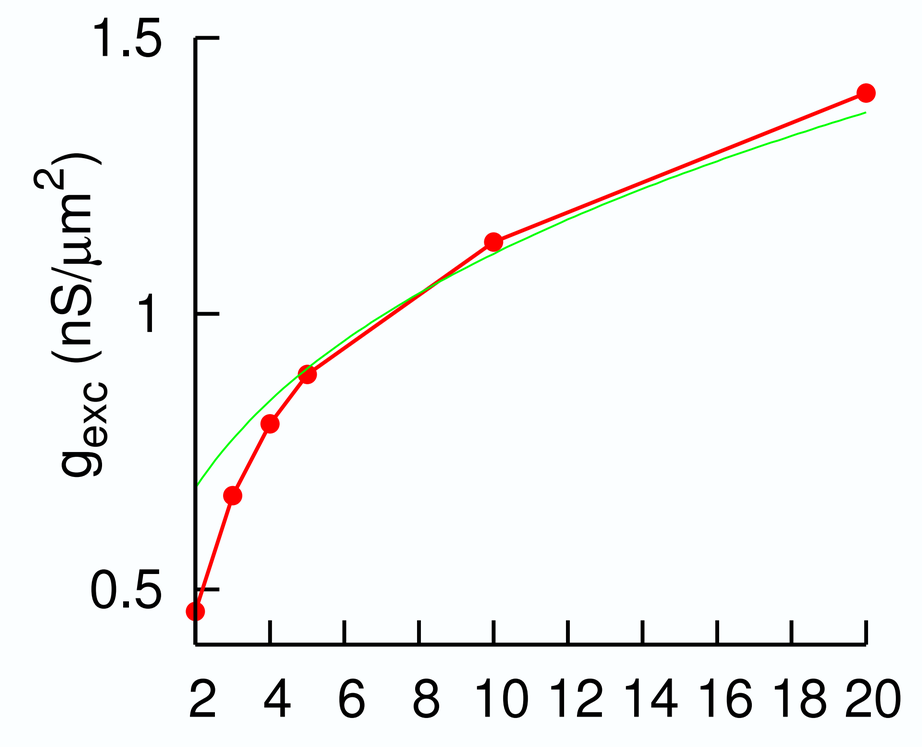
gexc (nS/μm2)

Dependence on Coupling

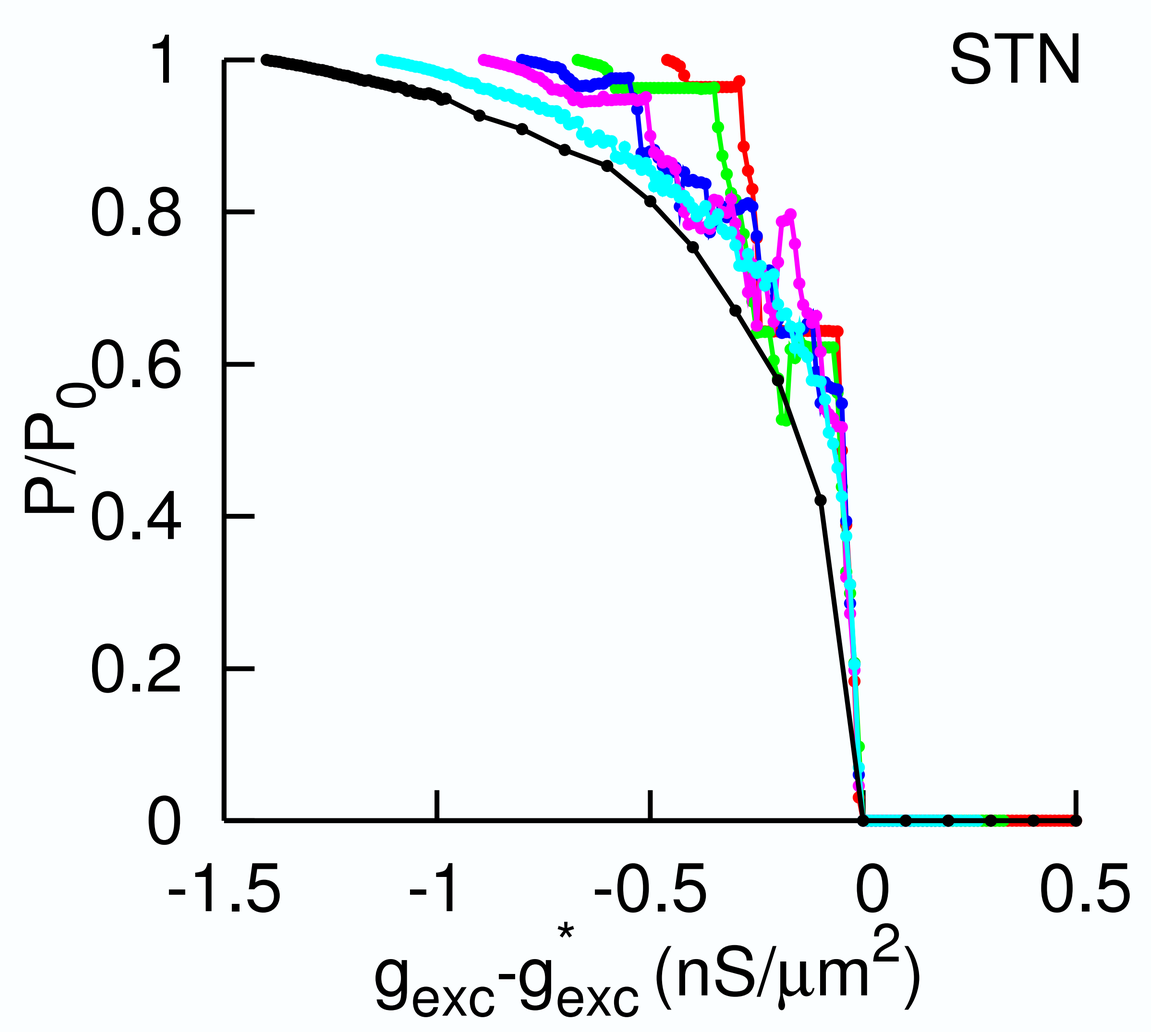
STN
P/P0
gexc-g*exc (nS/μm2)

A disorder parameter (P) that measures average asynchrony in the network is defined based on the ISI sequences. This parameter goes to zero in the synchronized state. By adjusting the critical strength for synchrony, we see that for all N (up to 20), the transition is approximately square root near the criticality.

Sparse Coupling

Synchrony in Sparsely Coupled Excitatory STN Neurons

Synchrony can occur even in very sparsely connected networks. Since the coupling has 1/n term, each synapse becomes stronger for sparse connectivity.

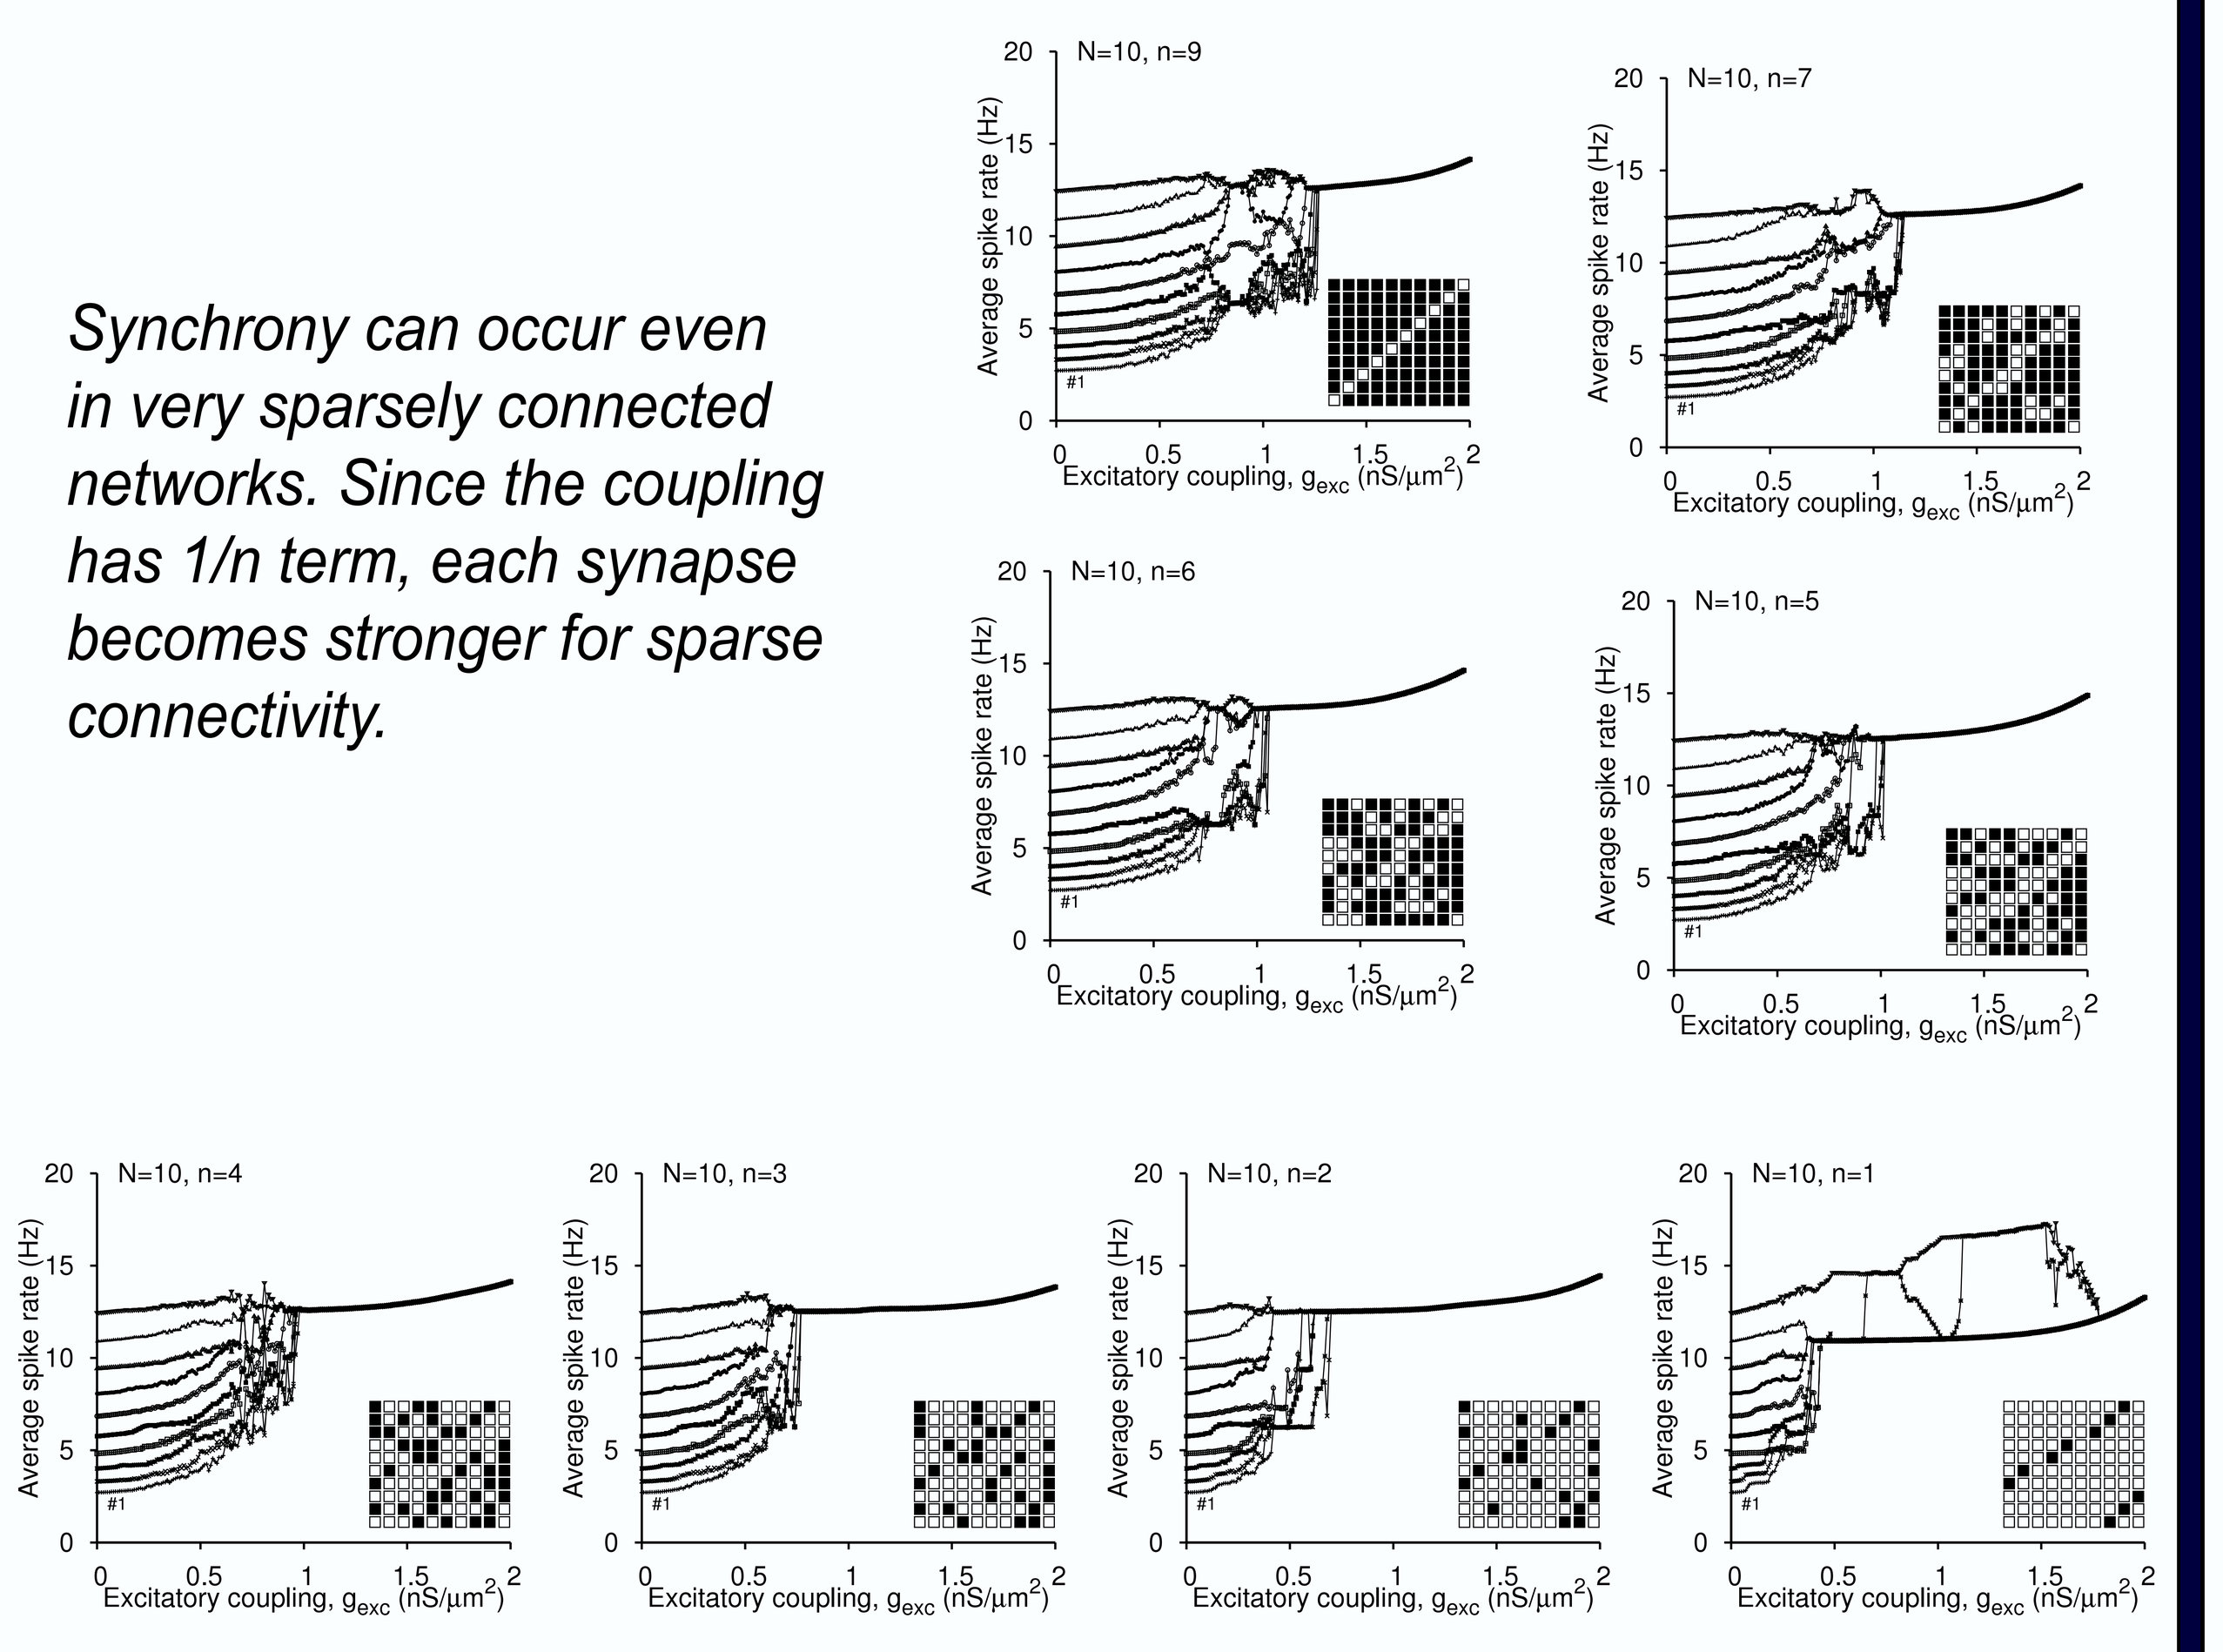
N=10, n=9
N=10, n=7
N=10, n=6
N=10, n=5
N=10, n=4
N=10, n=3
N=10, n=2
N=10, n=1
Average spike rate (Hz)
Excitatory coupling, gexc (nS/μm2)

Synchrony in Sparsely Coupled Inhibitory HH Neurons

In inhibitory networks, synchronized frequency is toward lower end of the frequency distribution. Sparseness in connectivity still retains considerable amount of synchrony.

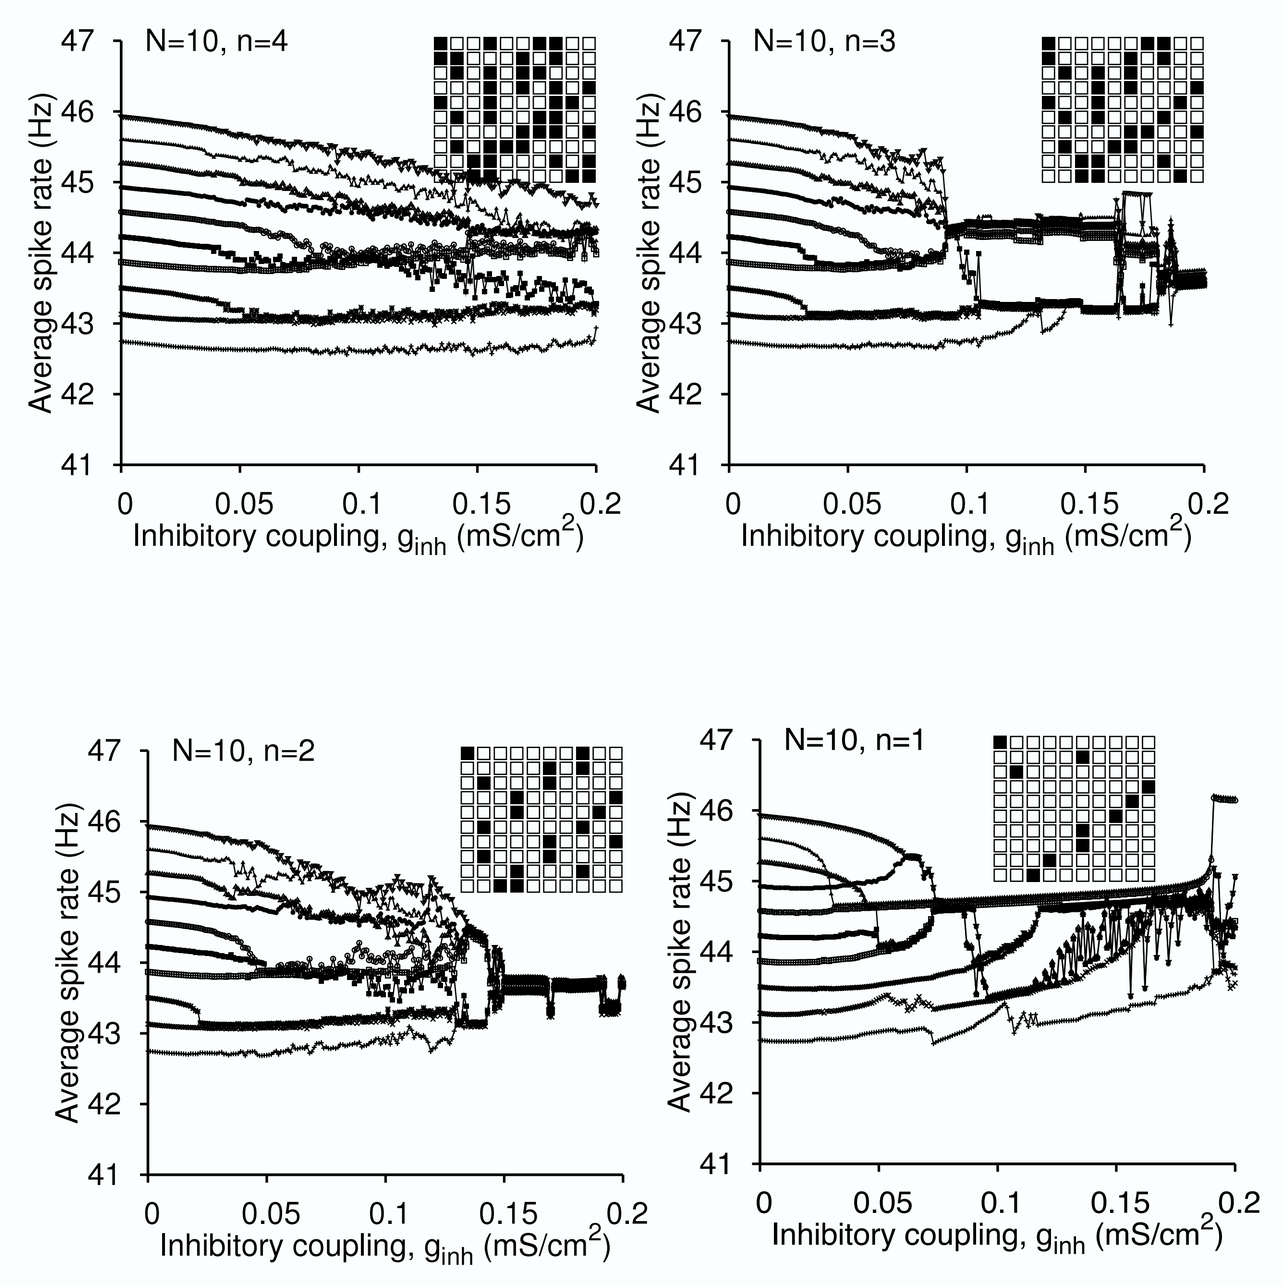
N=10, n=4
N=10, n=3
N=10, n=2
N=10, n=1
Average spike rate (Hz)
Inhibitory coupling, ginh (mS/cm2)

Loss of Synchrony due to Synaptic Input

Excitatory Network, STN

Spike skipping takes place in mutually excitatory STN neurons in response to weak input inhibition. At higher frequencies, rebound currents induce new and unpaired spikes.

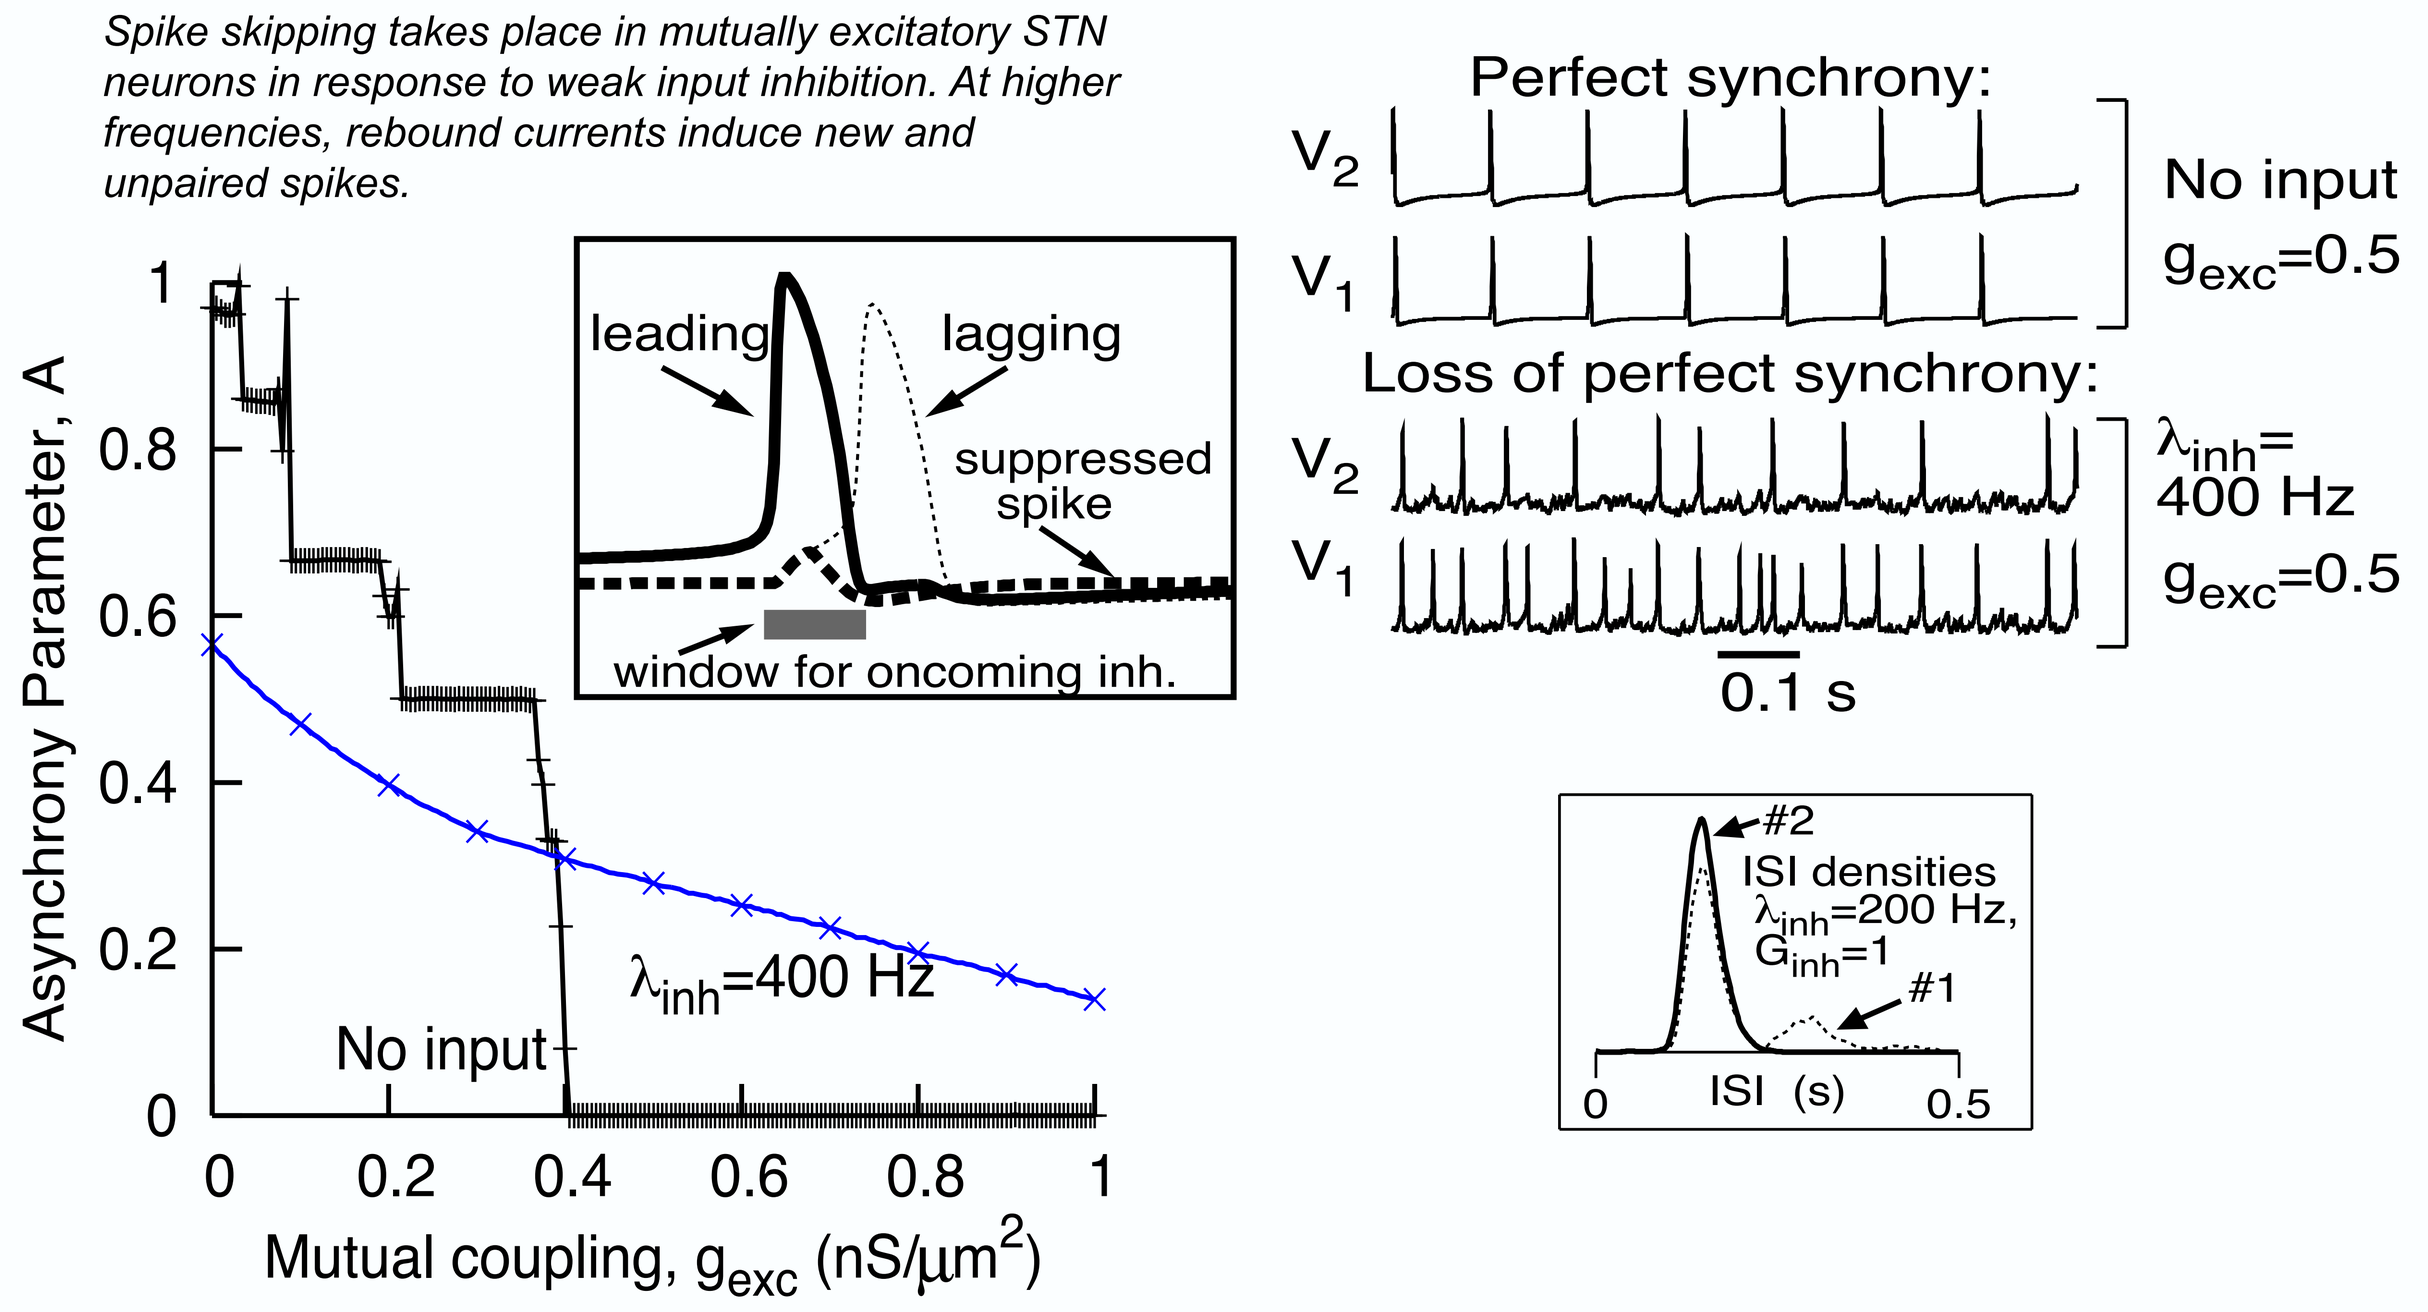
Asynchrony Parameter, A
leading
lagging
suppressed spike
window for oncoming inh.
λinh=400 Hz
No input
Mutual coupling, gexc (nS/μm2)
Perfect synchrony:
No input
gexc=0.5
Loss of perfect synchrony:
λinh=400 Hz
gexc=0.5
0.1 s
ISI densities
λinh=200 Hz, Ginh=1
ISI (s)

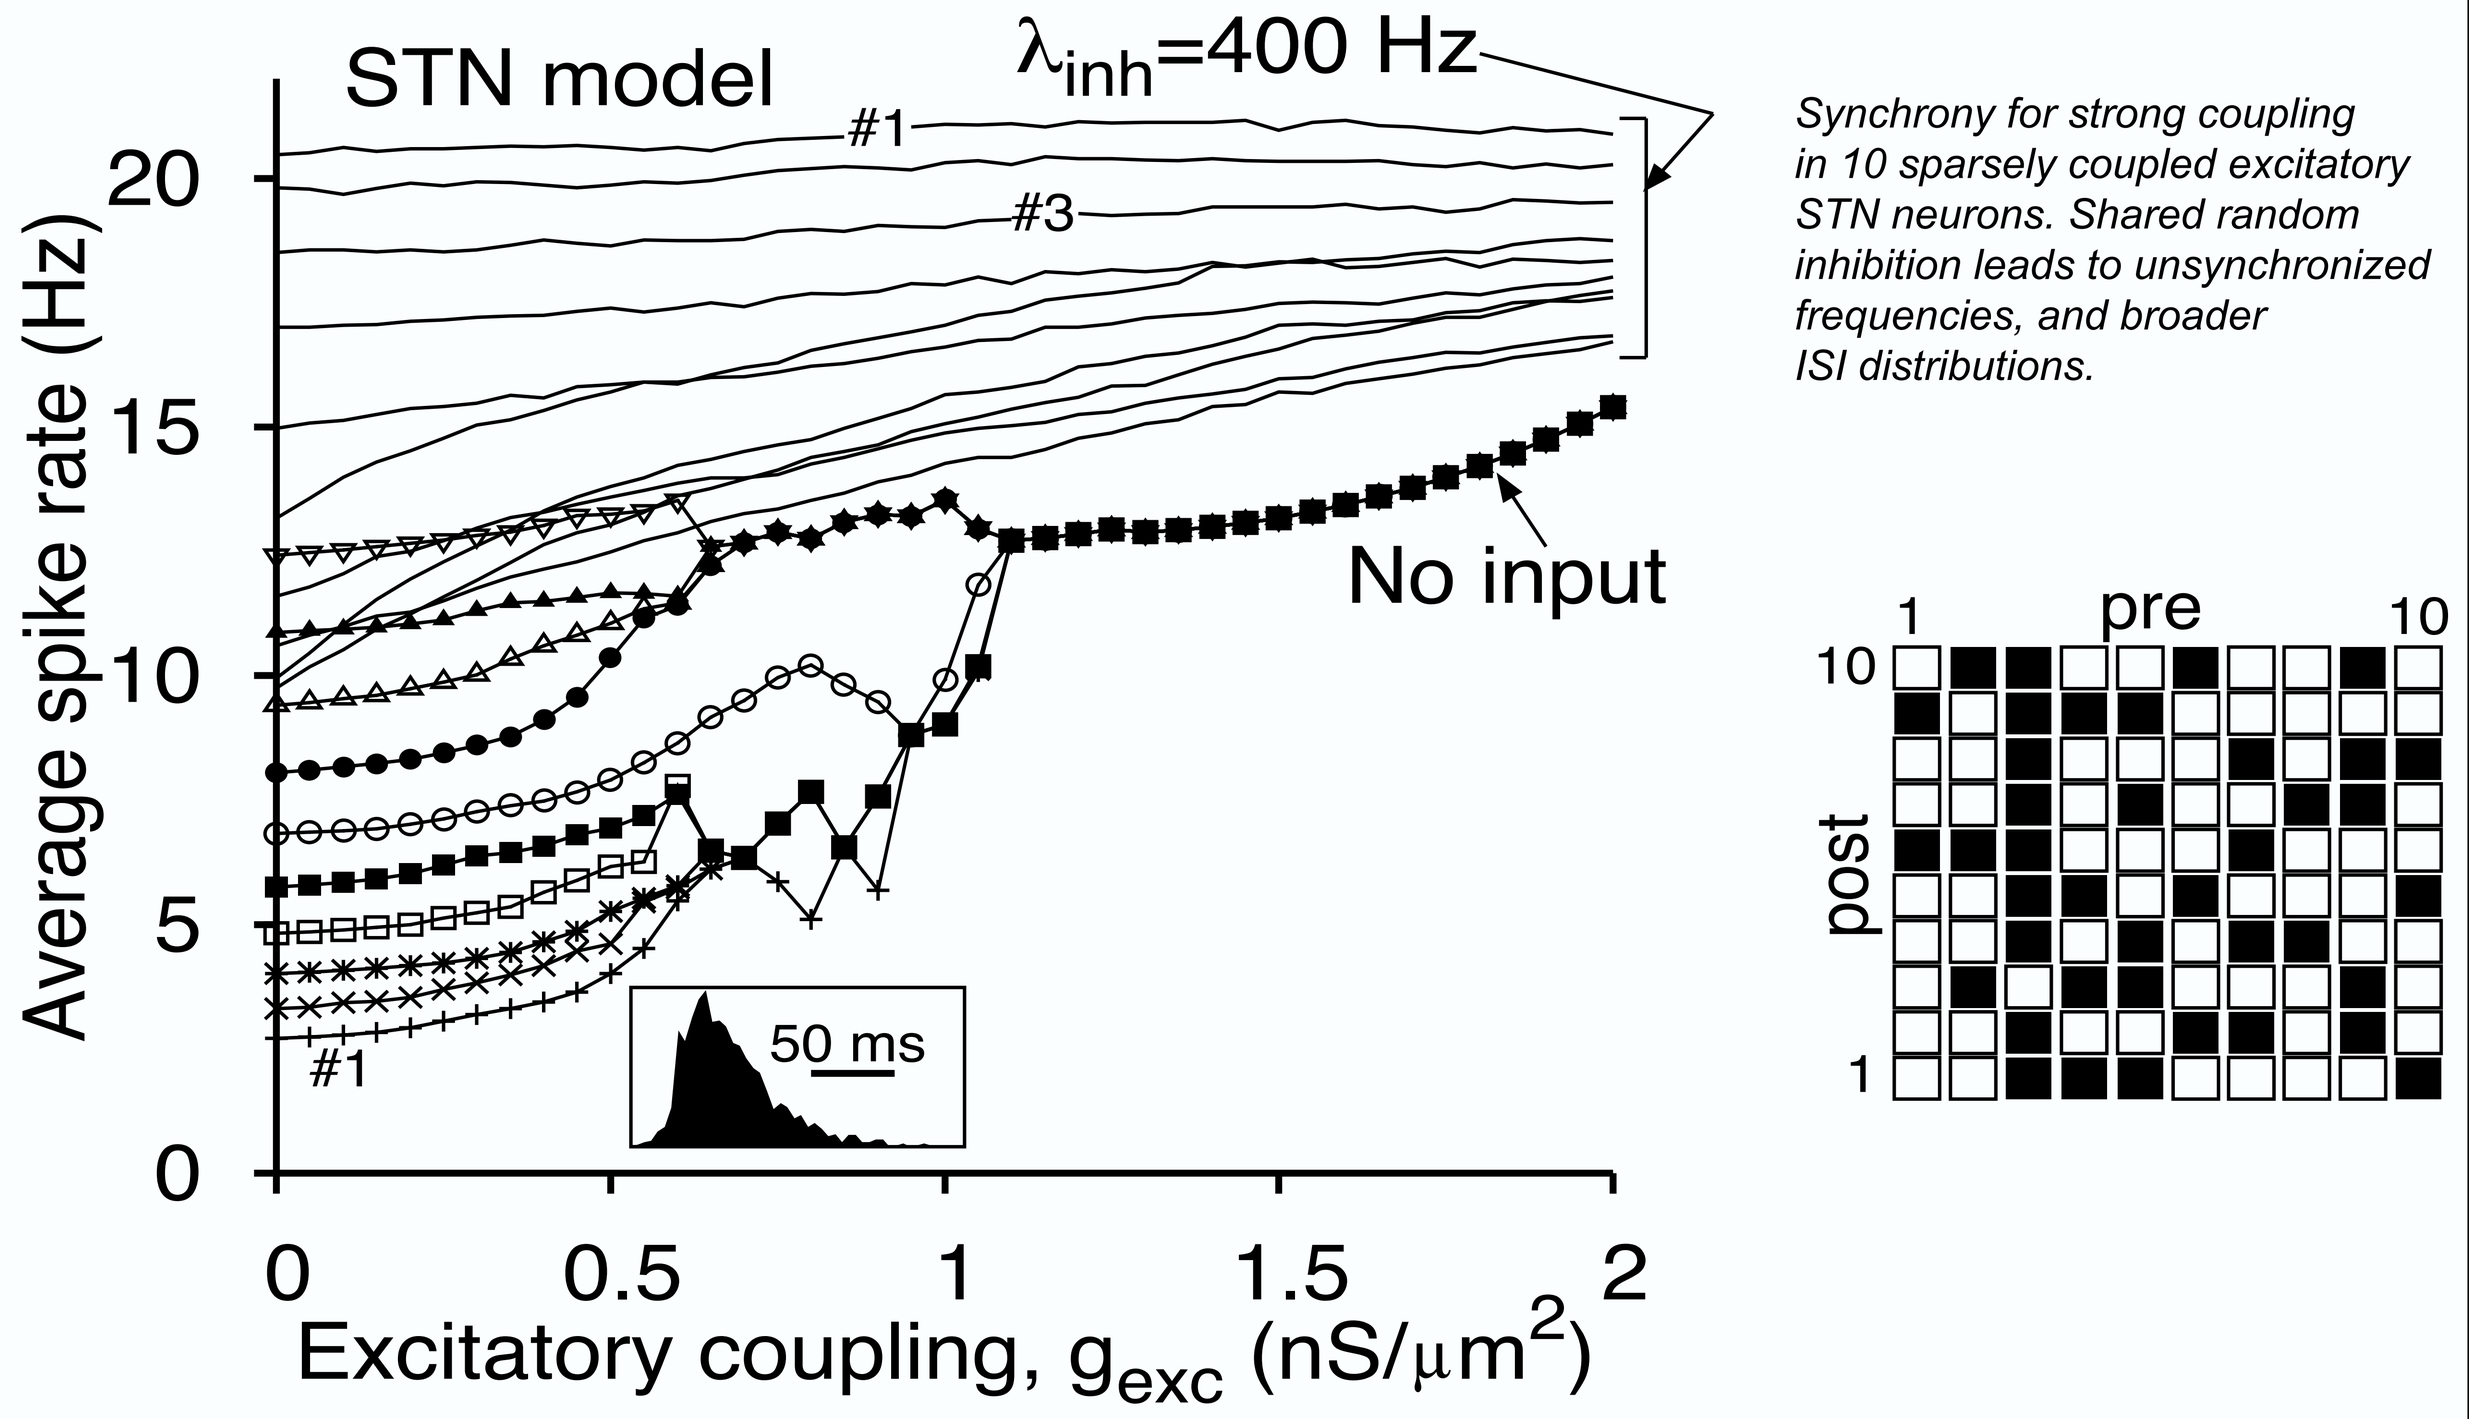
STN model
λinh=400 Hz
No input
50 ms
Average spike rate (Hz)
Excitatory coupling, gexc (nS/μm2)
pre
post

Synchrony for strong coupling in 10 sparsely coupled excitatory STN neurons. Shared random inhibition leads to unsynchronized frequencies, and broader ISI distributions.

Inhibitory Network, HH

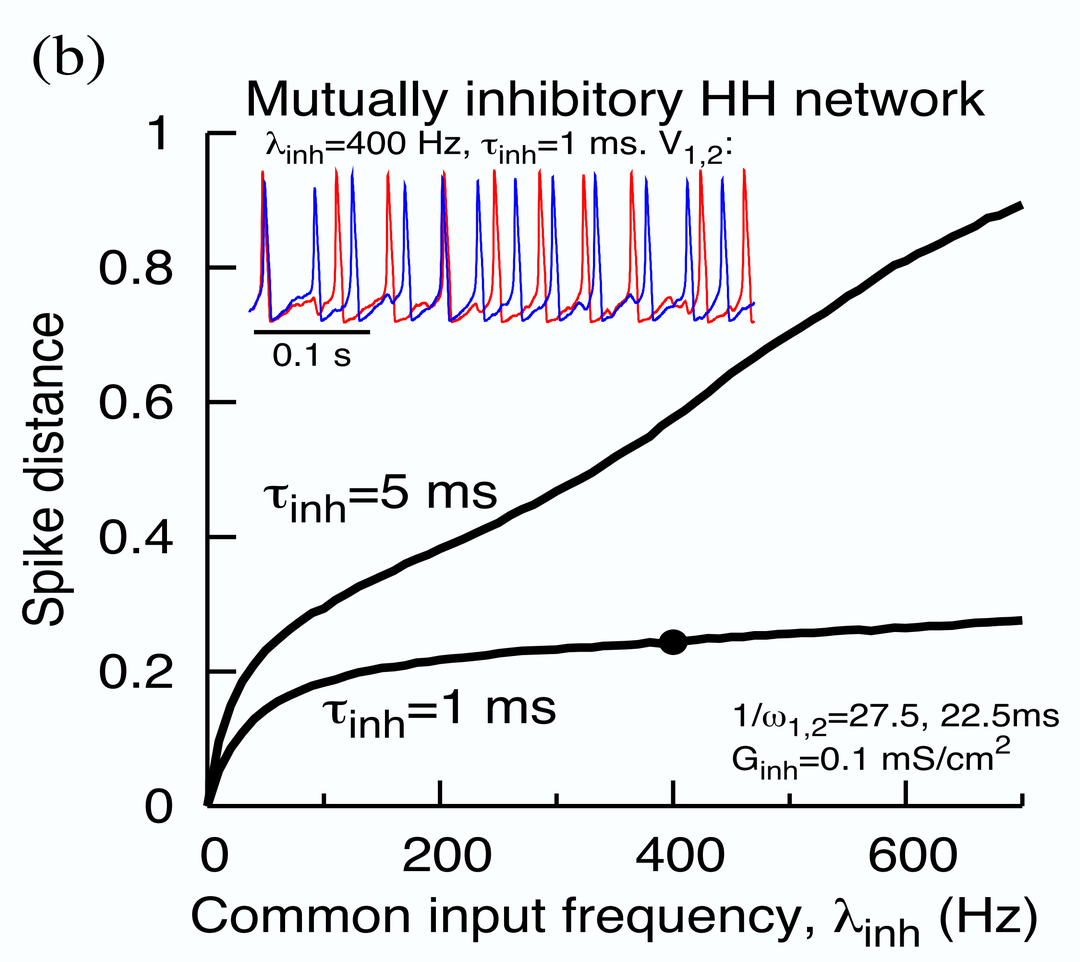
(b)
Mutually inhibitory HH network
λinh=400 Hz, τinh=1 ms. V1,2:
0.1 s
Spike distance
τinh=5 ms
τinh=1 ms
1/ω1,2=27.5, 22.5ms
Ginh=0.1 mS/cm2
Common input frequency, λinh (Hz)

Asynchrony arising between two mutually inhibiting HH neurons in response to shared inhibitory noise.

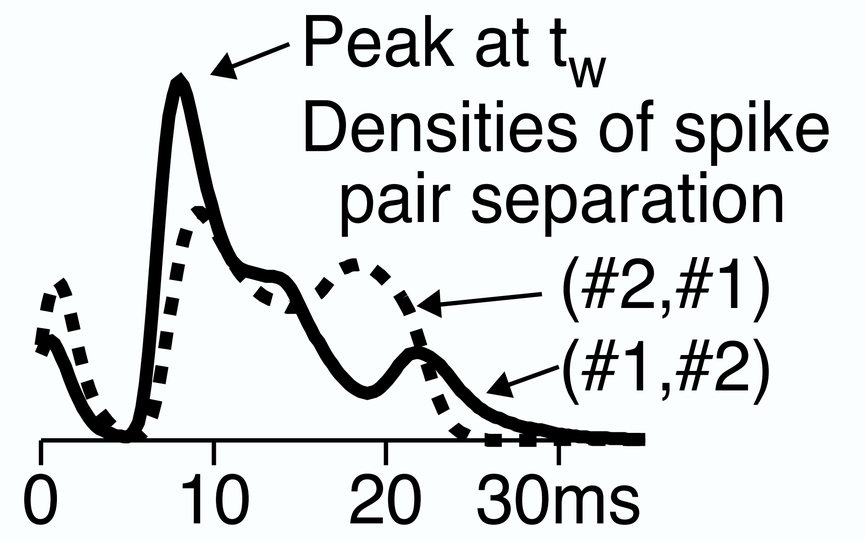
Peak at tw
Densities of spike pair separation
(#2,#1)
(#1,#2)
30ms

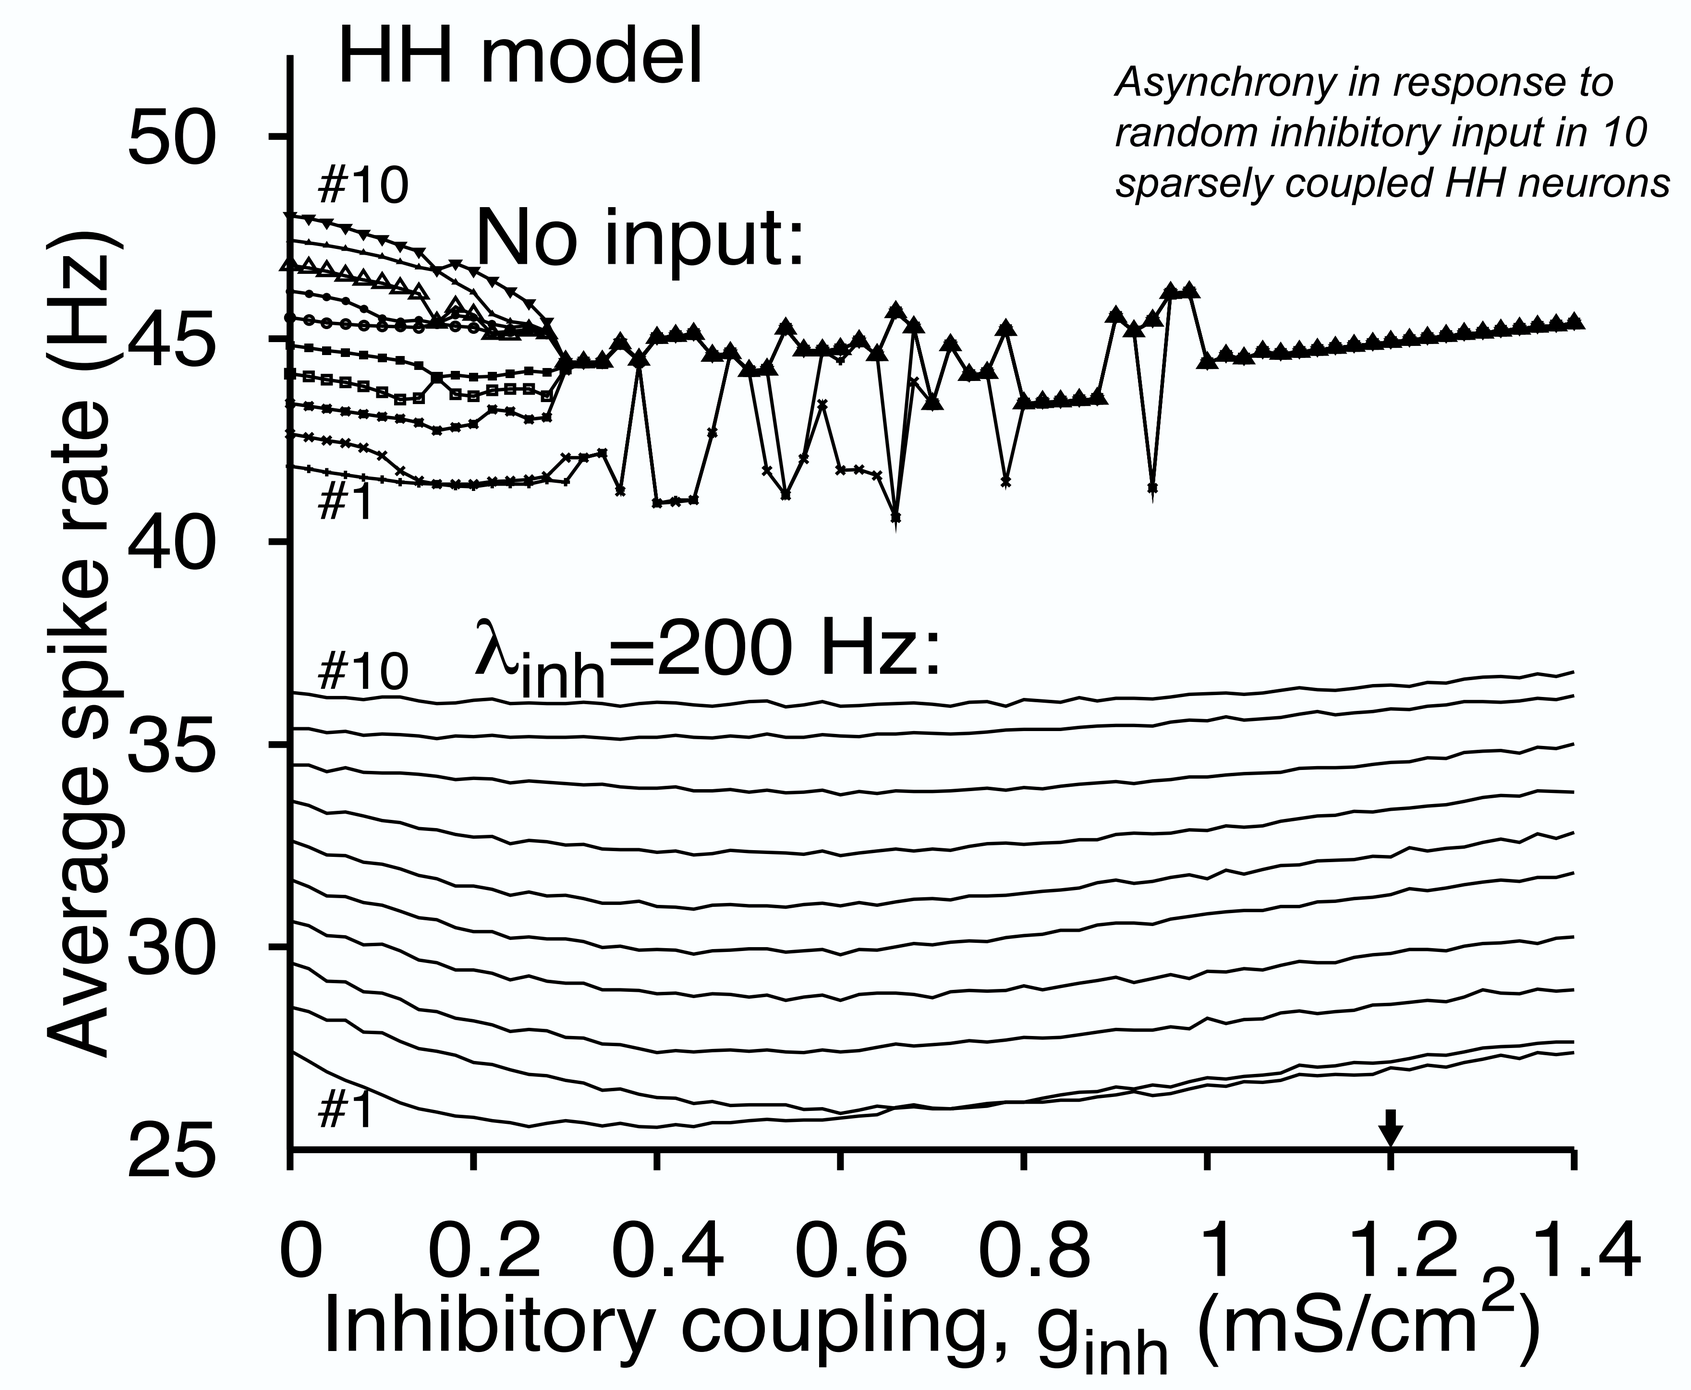
HH model
No input:
λinh=200 Hz:
Average spike rate (Hz)
Inhibitory coupling, ginh (mS/cm2)

Asynchrony in response to random inhibitory input in 10 sparsely coupled HH neurons

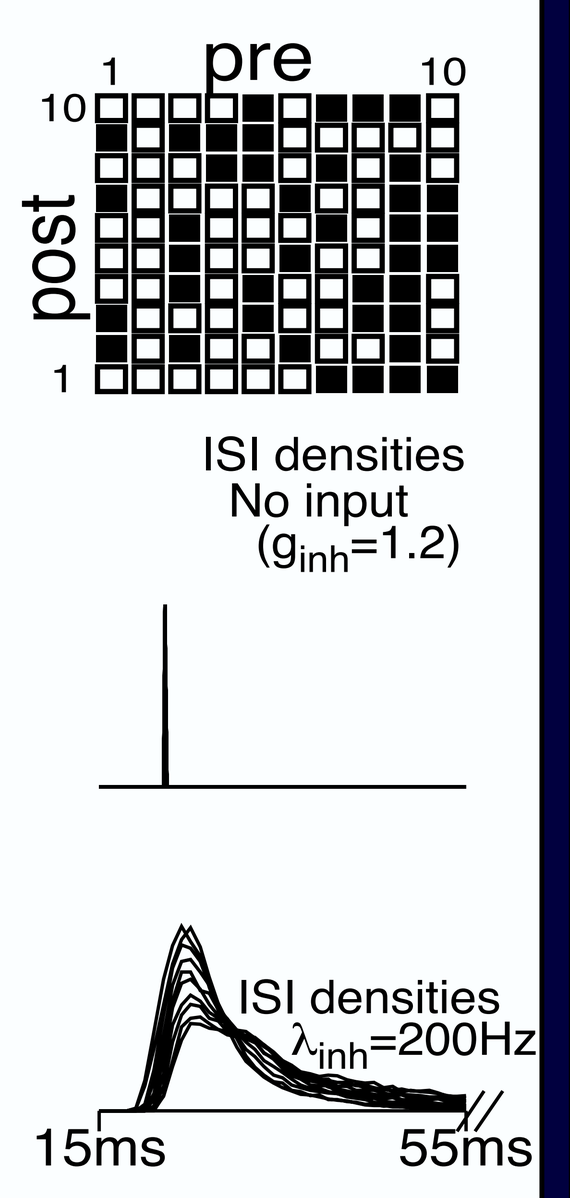
pre
post
ISI densities
No input
(ginh=1.2)
ISI densities
λinh=200Hz
15ms
55ms

Conclusions

Synchrony occurs in excitatory and inhibitory networks for all-to-all, as well as, for sparse coupling.

Synchrony is disrupted even for shared inputs.